Alpha-emitting radium-223: Two years follow up from a randomized phase II study in patients with bone metastases from hormone refractory prostate cancer

S. Nilsson[1], L. Franzén[2], C. Parker[3], B. Bolstad[4], T. Ramdahl[4], M. Thuresson[5] and Ø.S. Bruland[6]

[1]Karolinska University Hospital, Stockholm, Sweden; [2]Sundsvall Hospital, Sundsvall, Sweden; [3]Royal Marsden Hospital, Sutton, UK; [4]Algeta ASA, Oslo, Norway; [5]Statisticon AB, Uppsala, Sweden; [6]University of Oslo and The Radium Hospital, Oslo, Norway

BACKGROUND

The bone-seeking pharmaceutical Alpharadin® ($^{223}RaCl_2$) is under clinical development as a novel treatment for patients with skeletal metastases. Clinical studies have shown that toxicity is low, repeated dosing is feasible and seems to improve overall survival (1,2).
A pivotal phase-3 trial in patients with hormone refractory prostate cancer (HRPC) is currently recruiting.

METHODS AND TRIAL DESIGN

In a randomized phase 2 trial 64 HRPC patients with painful bone metastases received 4 monthly injections of Alpharadin or placebo as an adjuvant to external beam radiotherapy (EBRT).
A 4.5 months difference in survival was observed at 18 months follow up (2). Here we report 24 months follow up data on survival, long term toxicity, sub-group analyses based on disease status at inclusion and pre-treatment EBRT.

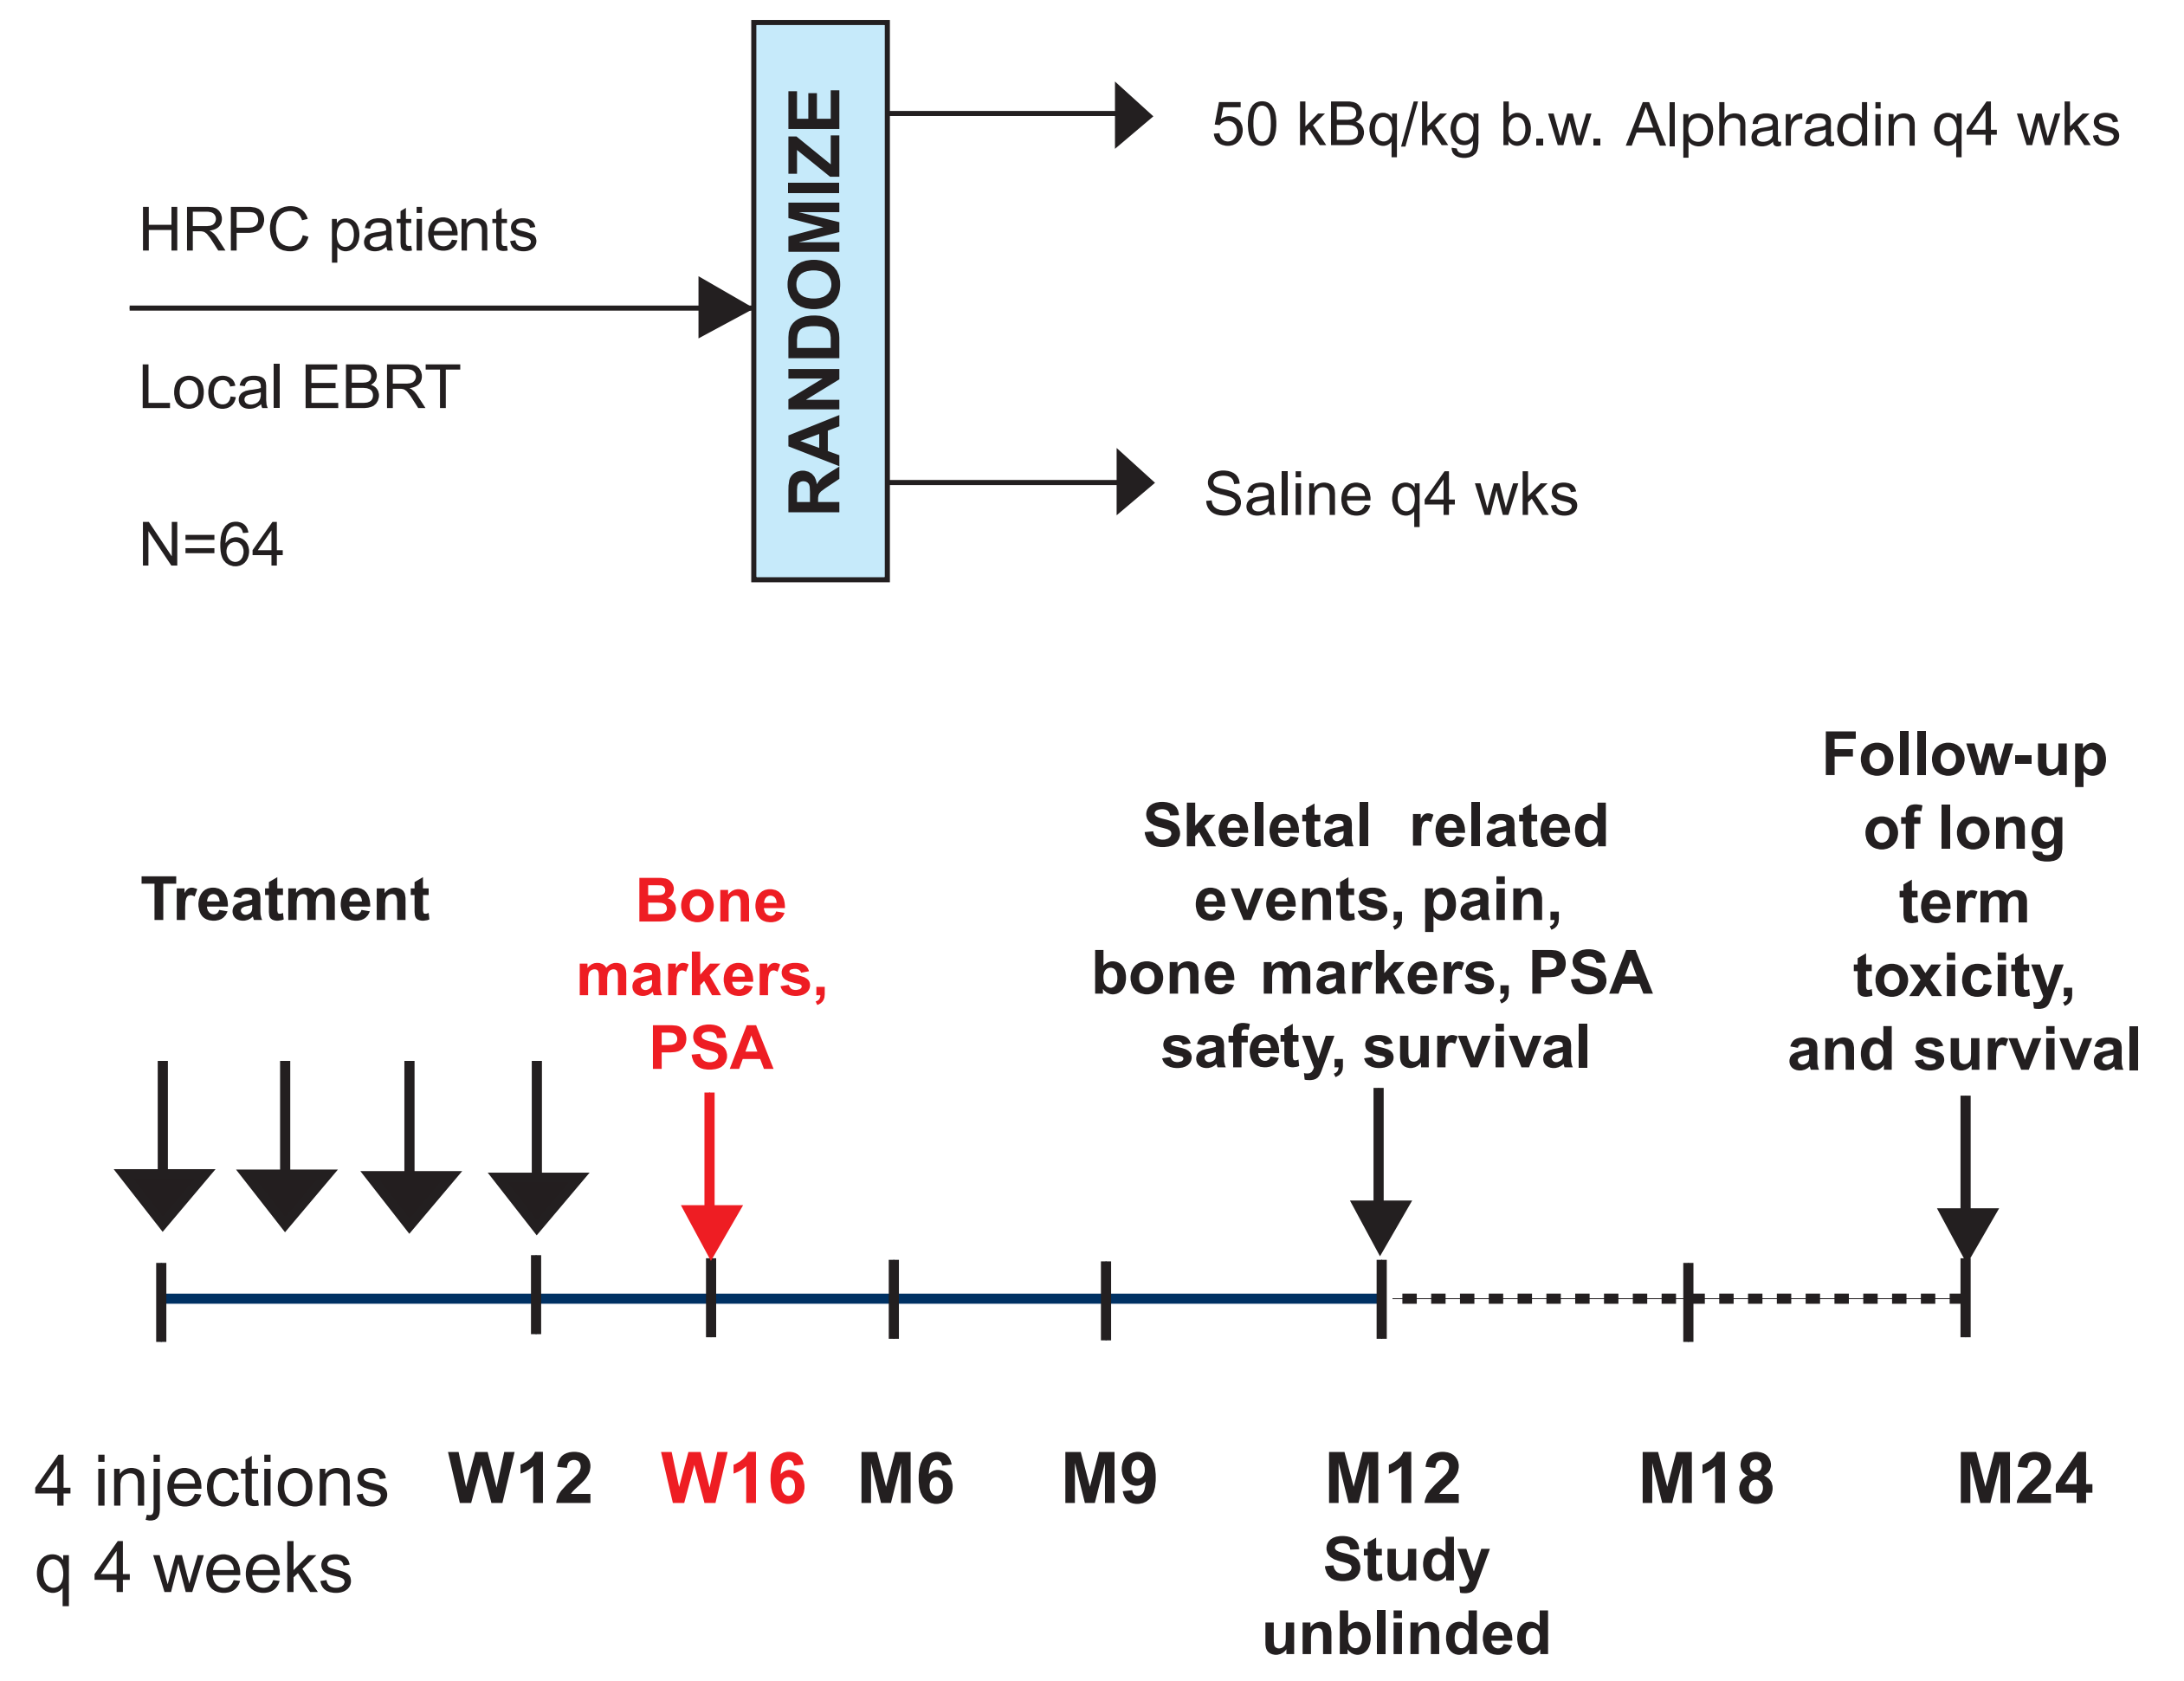
HRPC patients
Local EBRT
N=64
RANDOMIZE
50 kBq/kg b.w. Alpharadin q4 wks
Saline q4 wks
Treatment
Bone markers, PSA
Skeletal related events, pain, bone markers, PSA safety, survival
Follow-up of long term toxicity, and survival
4 injections q 4 weeks
W12
W16
M6
M9
M12 Study unblinded
M18
M24

MAIN ELIGIBILITY CRITERIA

- Confirmed hormone refractory prostate cancer with painful skeletal metastases
- Referred to palliative external radiotherapy for skeletal metastases
- No other currently active malignancy or known metastases to other organ than skeleton

RESULTS

TWO YEAR SURVIVAL

At 24 months, ten patients (30%) that received Alpharadin were alive and four (13%) in the placebo-group. Median survival was 65 weeks compared with 46 weeks, respectively (ITT). Hazard ratio adjusted for baseline covariates was 2.10 (95% CI; 1.14-3.88; p=0.017, Cox regression).

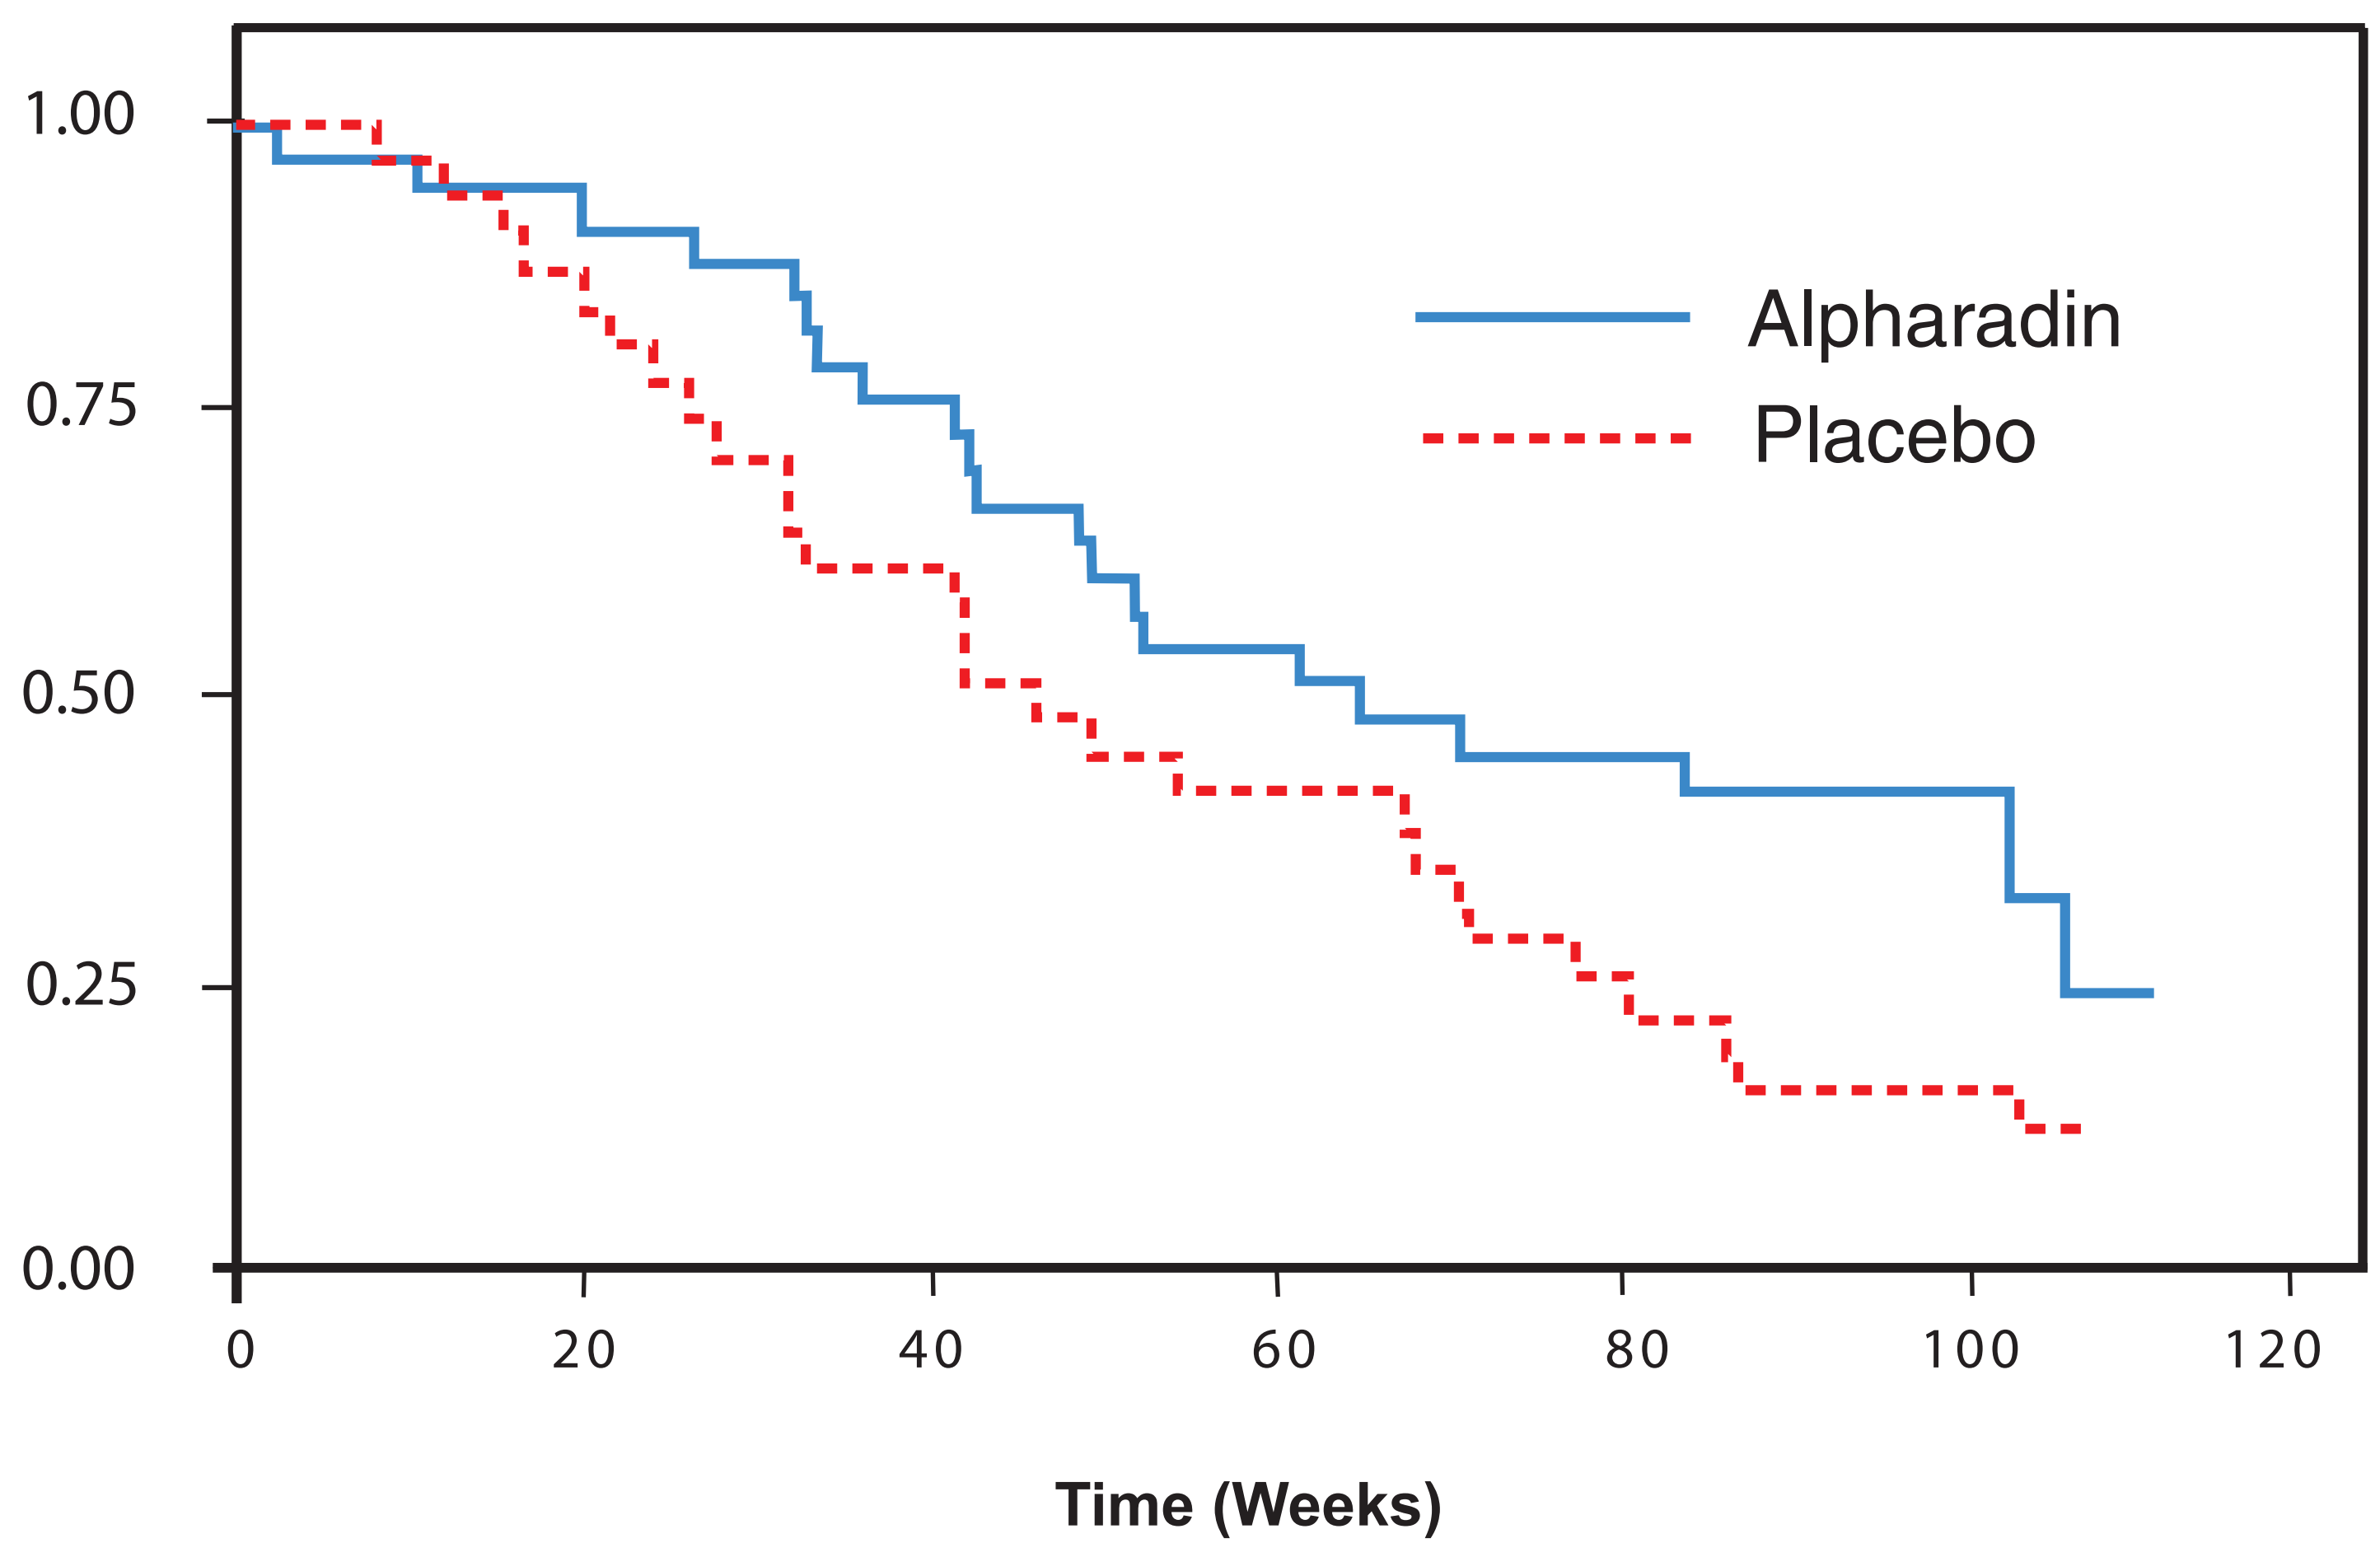
1.00
0.75
0.50
0.25
0.00
0
20
40
60
80
100
120
Alpharadin
Placebo
Time (Weeks)

SURVIVAL VS. EOD

The median survival was more than 40% longer in the Alpharadin group at all levels of extent of disease (EOD = number of "hot-spots": <6; 6-20; >20; super-scan). The largest absolute difference occurred in patients with lowest EOD; 107 weeks for Alpharadin and 68 weeks for placebo.

	Alpharadin			Placebo		
EOD	N	N events	Median (week)	N	N events	Median (week)
<6	12	6	106.7	7	5	68.3
6-20	10	7	77.4	13	12	54.3
>20	10	9	42.5	10	9	30.0
Superscan	1	1	61.6	1	1	24.6

SURVIVAL VS. NUMBER OF INJECTIONS

For all patients who received four injections (28 patients in Alpharadin, 21 in placebo), the median survival was 93 weeks for Alpharadin and 49 weeks for placebo (p = 0.0427, Log-rank).

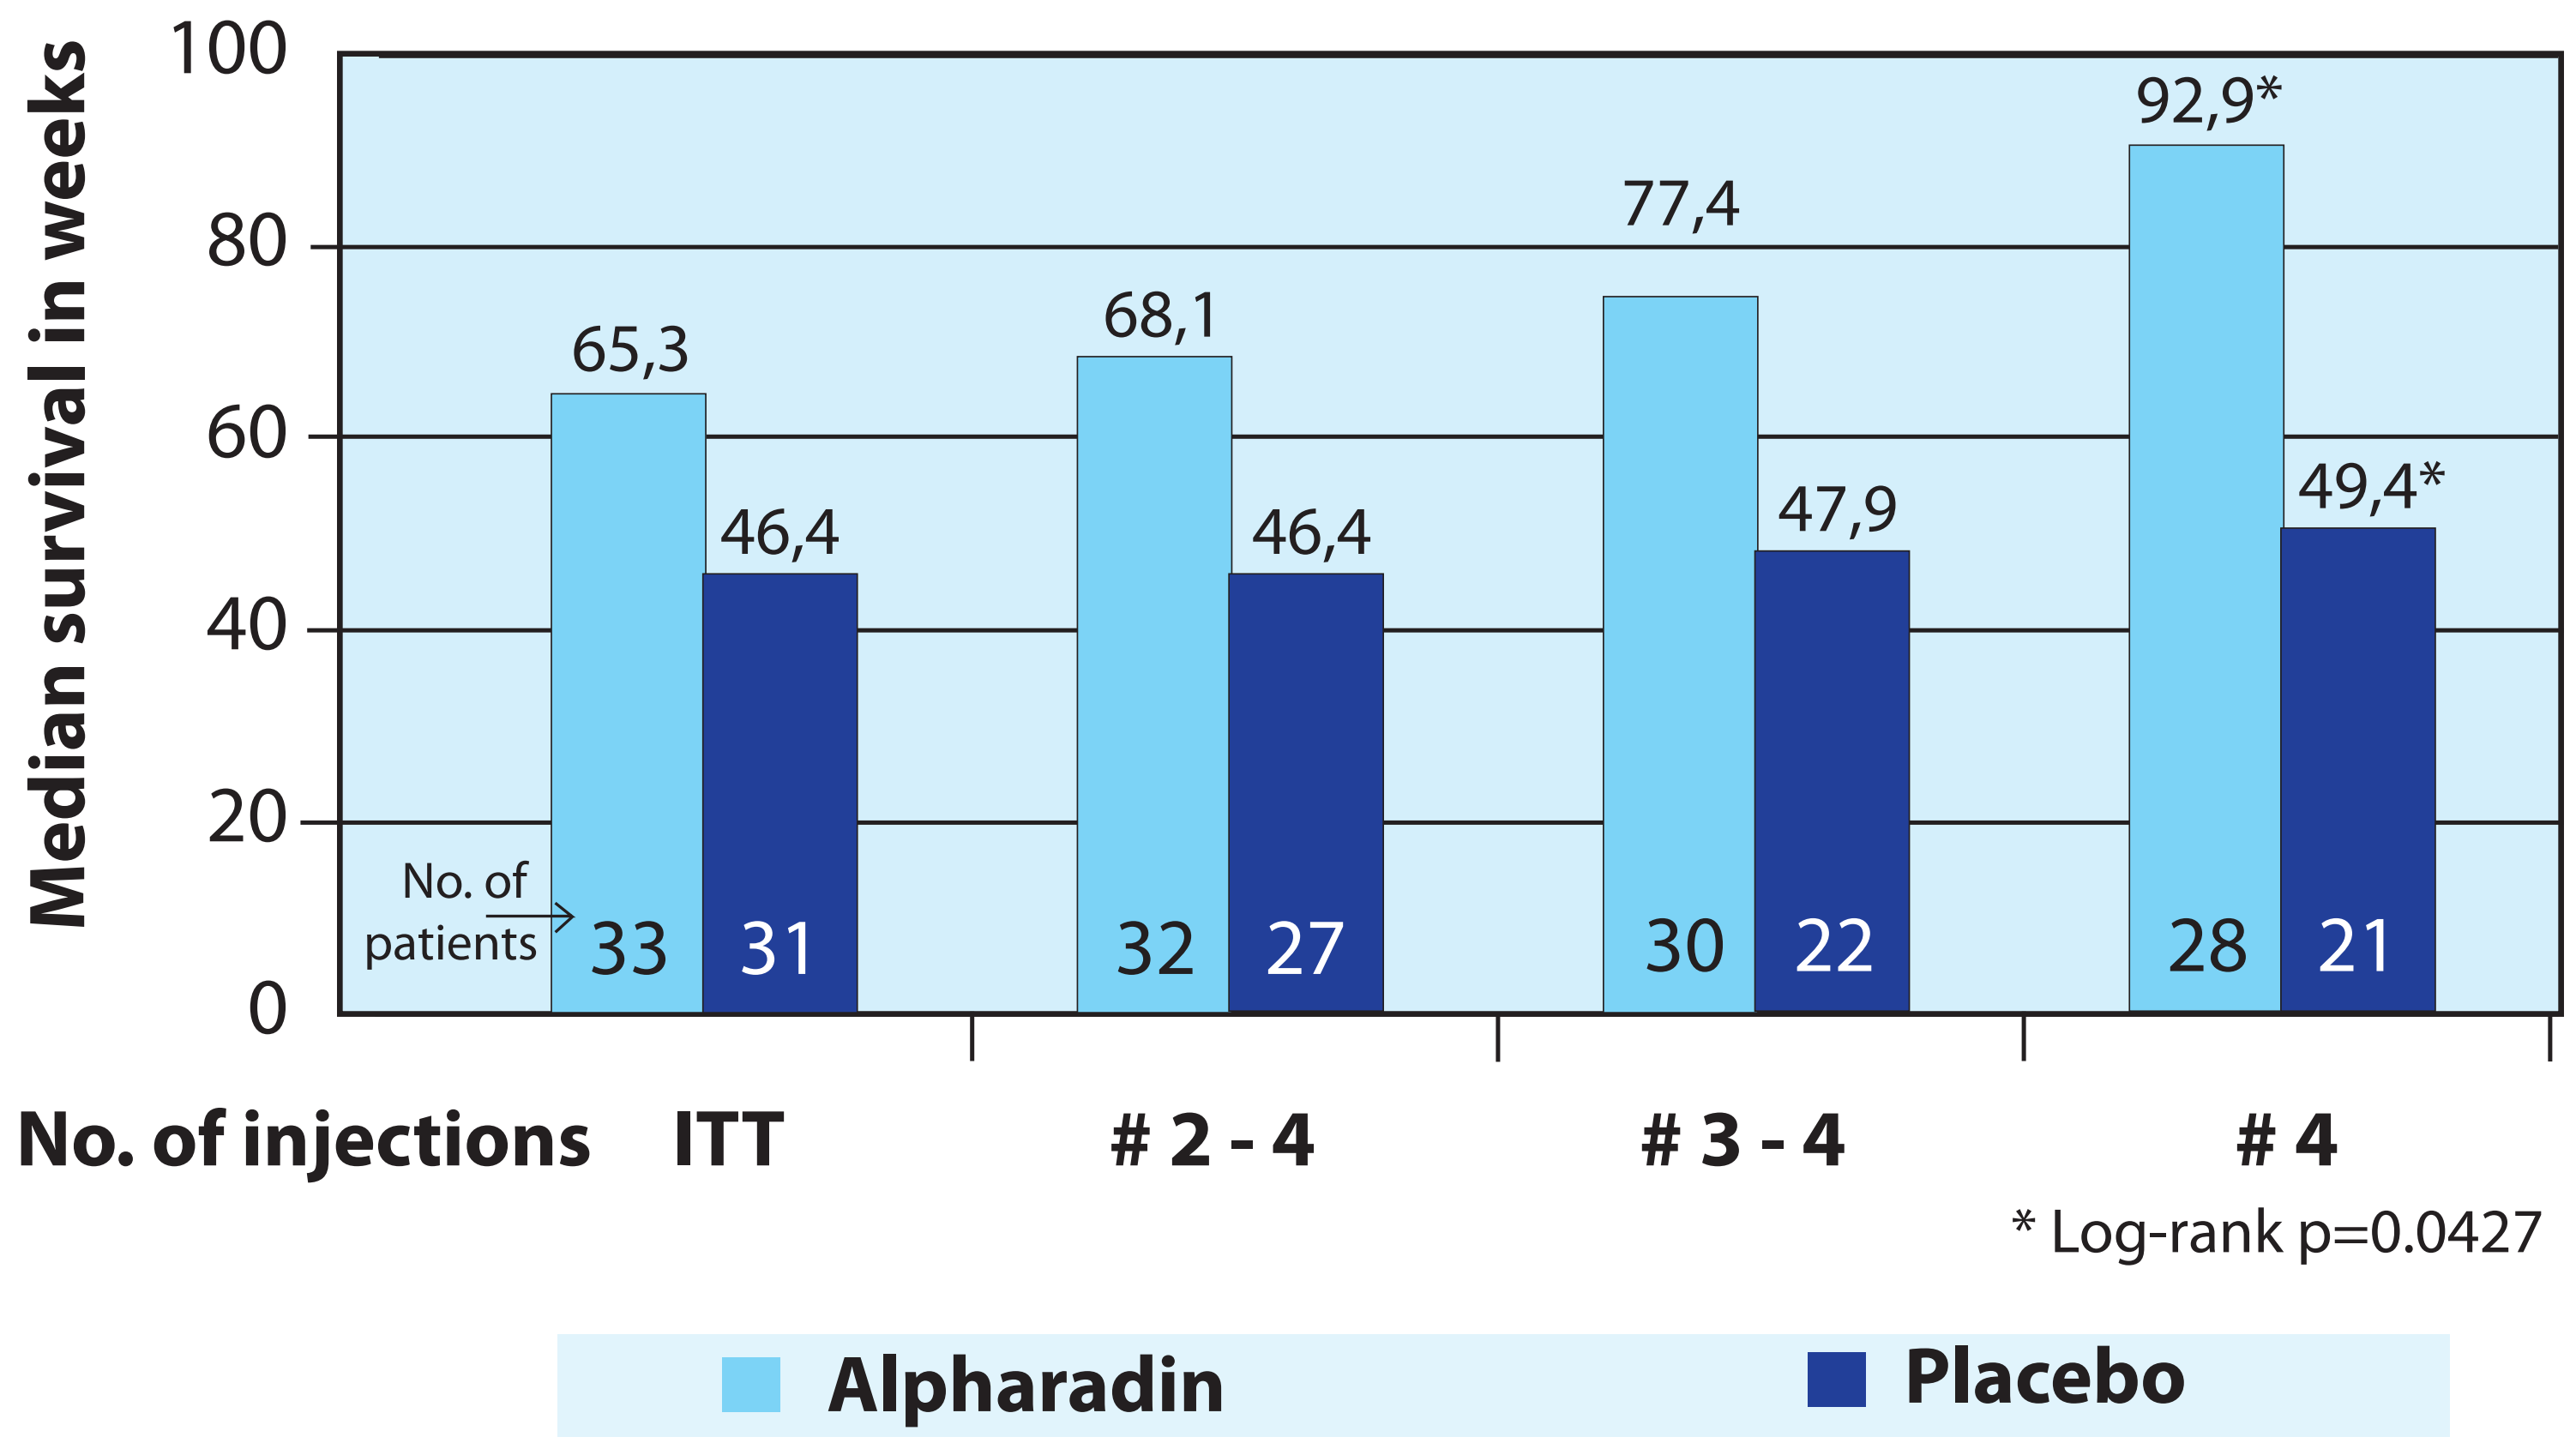
Median survival in weeks
100
80
60
40
20
0
65,3
46,4
68,1
46,4
77,4
47,9
92,9*
49,4*
No. of patients
33
31
32
27
30
22
28
21
No. of injections
ITT
2 - 4
3 - 4
4
* Log-rank p=0.0427
Alpharadin
Placebo

OTHER SIGNIFICANT FINDINGS

In a composite analysis of patients with Hb >110 g/dl, bone-ALP <800 ng/ml, and PSA <1,000 ng/ml the median survival was 102 versus 49 weeks in the two groups (p= 0.029, Log-rank).
Patients with body mass index above 23,0 (27 patients in Alpharadin and 25 in placebo) had a median survival of 102 and 54 weeks, respectively (p=0.035, Log-rank). The administered EBRT at baseline was comparable between the two groups with no difference in the use of single dose versus fractionated EBRT.

NO LONG TERM TOXICITY

No long term haematological toxicity or any cases of leukaemia, myelodysplastic syndrome, aplastic anaemia or bone sarcoma were reported.

CONCLUSION

The therapeutic benefit seems to be greater for more fit patients than for those with extensive skeletal involvement. However, the relative improvement in survival was maintained irrespective of extent of disease at baseline. A benign side effect profile was documented following repeated Alpharadin treatment.

REFERENCES

1) Nilsson S et al., Clin Cancer Res. 2005;11 (12): 4451-4459
2) Nilsson S et al., Lancet Oncology 2007; 8 (7): 587-594