Abstract 4620

Alkaline Phosphatase (ALP) Normalization and Overall Survival in Patients With Bone Metastases From Castration-Resistant Prostate Cancer (CRPC) Treated With Radium-223 (Alpharadin™)

S. Nilsson,[1] C. G. O'Bryan-Tear,[2] B. Bolstad,[3] A. Lokna,[2] C. Parker[4]

[1]Karolinska University Hospital, Stockholm, Sweden; [2]Algeta ASA, Oslo, Norway; [3]Nordic Nanovector AS, Oslo, Norway; [4]Royal Marsden Hospital, Sutton, United Kingdom

BACKGROUND

- Bone metastases are the main cause of disability and death in patients with castration-resistant prostate cancer (CRPC)[1]
- Radium-223 chloride (Alpharadin™) is a first-in-class alpha-pharmaceutical with a potent and highly targeted antitumor effect on bone metastases[2] and a highly tolerable side effect profile[3]
- Bone-targeted radium-223 emits alpha-particles with an ultra-short range of 2-10 cell diameters, generating highly localized and intense radiation zones.[2] The high-energy alpha-particle radiation induces lethal double-stranded DNA breaks,[4] resulting in a potent and highly localized cytotoxic effect in the target areas containing metastatic cancer cells. The short path length of the alpha-particles also ensures that toxicity to adjacent healthy tissue and particularly the bone marrow is kept to a minimum[2]
- In the placebo-controlled phase II study (BC1-02), radium-223 showed a statistically significant improvement in overall survival (OS), compared with placebo,[3,5] and a consistent improvement in pain relief[6] and disease-related biomarkers such as prostate-specific antigen (PSA) and bone alkaline phosphatase (ALP)[3,6,7]
- Baseline ALP values have been shown to be prognostic for survival in men with metastatic prostate cancer[8]; however, the relationship between a change in baseline ALP in patients receiving treatment and the subsequent effect on OS has not been completely determined
- In a retrospective analysis of the TAX327 trial, ALP normalization at 90 days occurred in 26% of patients treated with docetaxel or mitoxantrone and correlated with better survival[9]
- Since radium-223 has been shown to have a profound effect on serum markers of bone metabolism, including ALP,[3,6,7] an exploratory analysis was conducted to assess whether the effect of radium-223 on ALP is associated with improved survival

METHODS AND STUDY DESIGN

- Patients with CRPC and bone metastases were treated in 2 randomized phase II studies. BC1-02 was a placebo-controlled study of radium-223 (50 kBq/kg) every 4 weeks for 12 weeks (**Figure 1**). BC1-04 evaluated radium-223 (25, 50, or 80 kBq/kg) every 6 weeks for 12 weeks (**Figure 2**)
- The trials were designed to assess the safety (eg, hematology and adverse events [AEs]) and efficacy (eg, survival, pain relief, and changes in PSA and ALP) of radium-223 in patients with CRPC and bone metastases
- Ad hoc analyses were conducted to determine OS for patients with above-normal baseline total ALP values. The intent of this analysis was to compare the difference in OS between patients with normalized versus nonnormalized ALP values after treatment (ie, radium-223 or placebo)
 - All patients were classified according to their baseline total ALP values; values > 128 U/L were classified as above normal, and values ≤ 128 U/L were identified as normal
 - Patients with elevated baseline ALP values were evaluated to determine if they had a value below normal (≤ 128 U/L) after treatment
 - Patients with ALP values decreasing to ≤ 128 U/L at week 12 (BC1-02) or week 13 (BC1-04) were defined as normalized, and those with ALP values remaining > 128 U/L were defined as nonnormalized
 - OS was compared for normalized versus nonnormalized patients using a log-rank analysis

Figure 1. Design of the placebo-controlled, phase II BC1-02 study

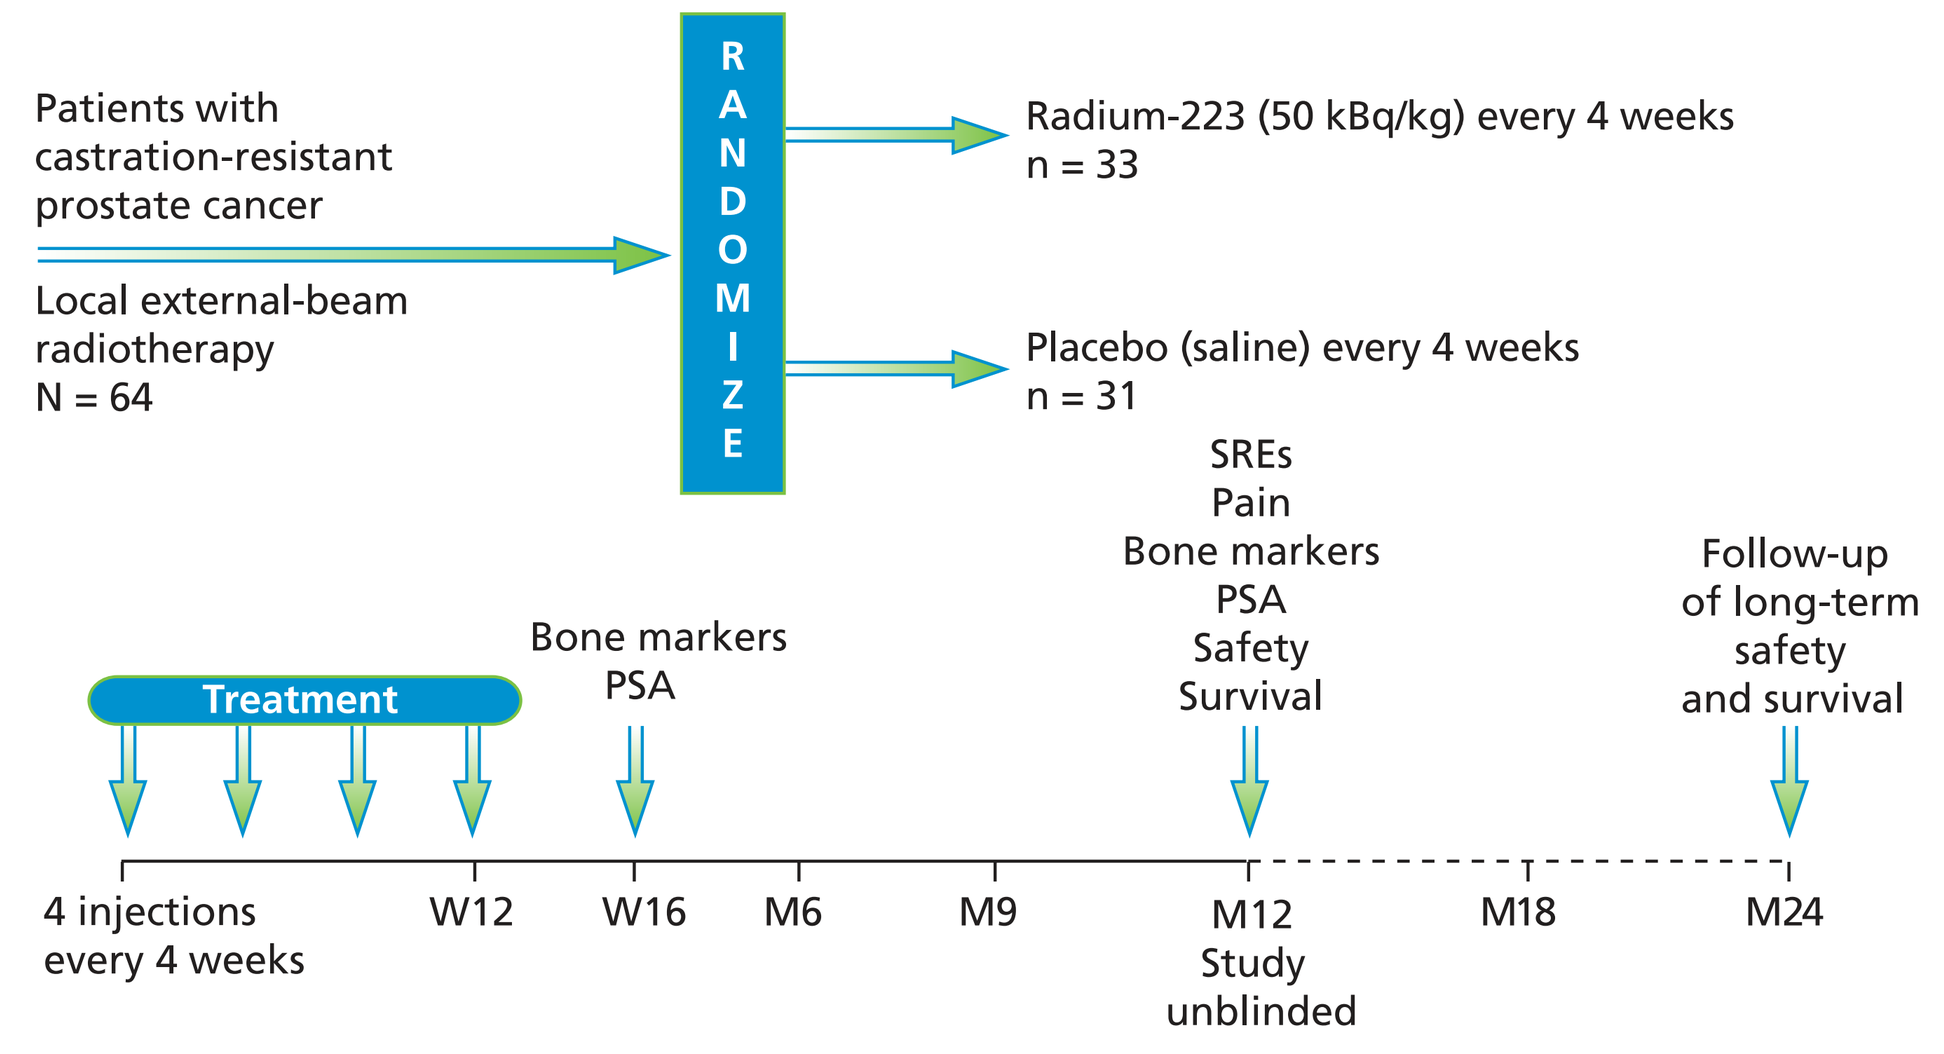
Patients with castration-resistant prostate cancer
Local external-beam radiotherapy
N = 64
RANDOMIZE
Radium-223 (50 kBq/kg) every 4 weeks
n = 33
Placebo (saline) every 4 weeks
n = 31
Treatment
Bone markers
PSA
SREs
Pain
Bone markers
PSA
Safety
Survival
Follow-up of long-term safety and survival
4 injections every 4 weeks
W12
W16
M6
M9
M12
Study unblinded
M18
M24

Figure 2. Design of the phase II BC1-04 study

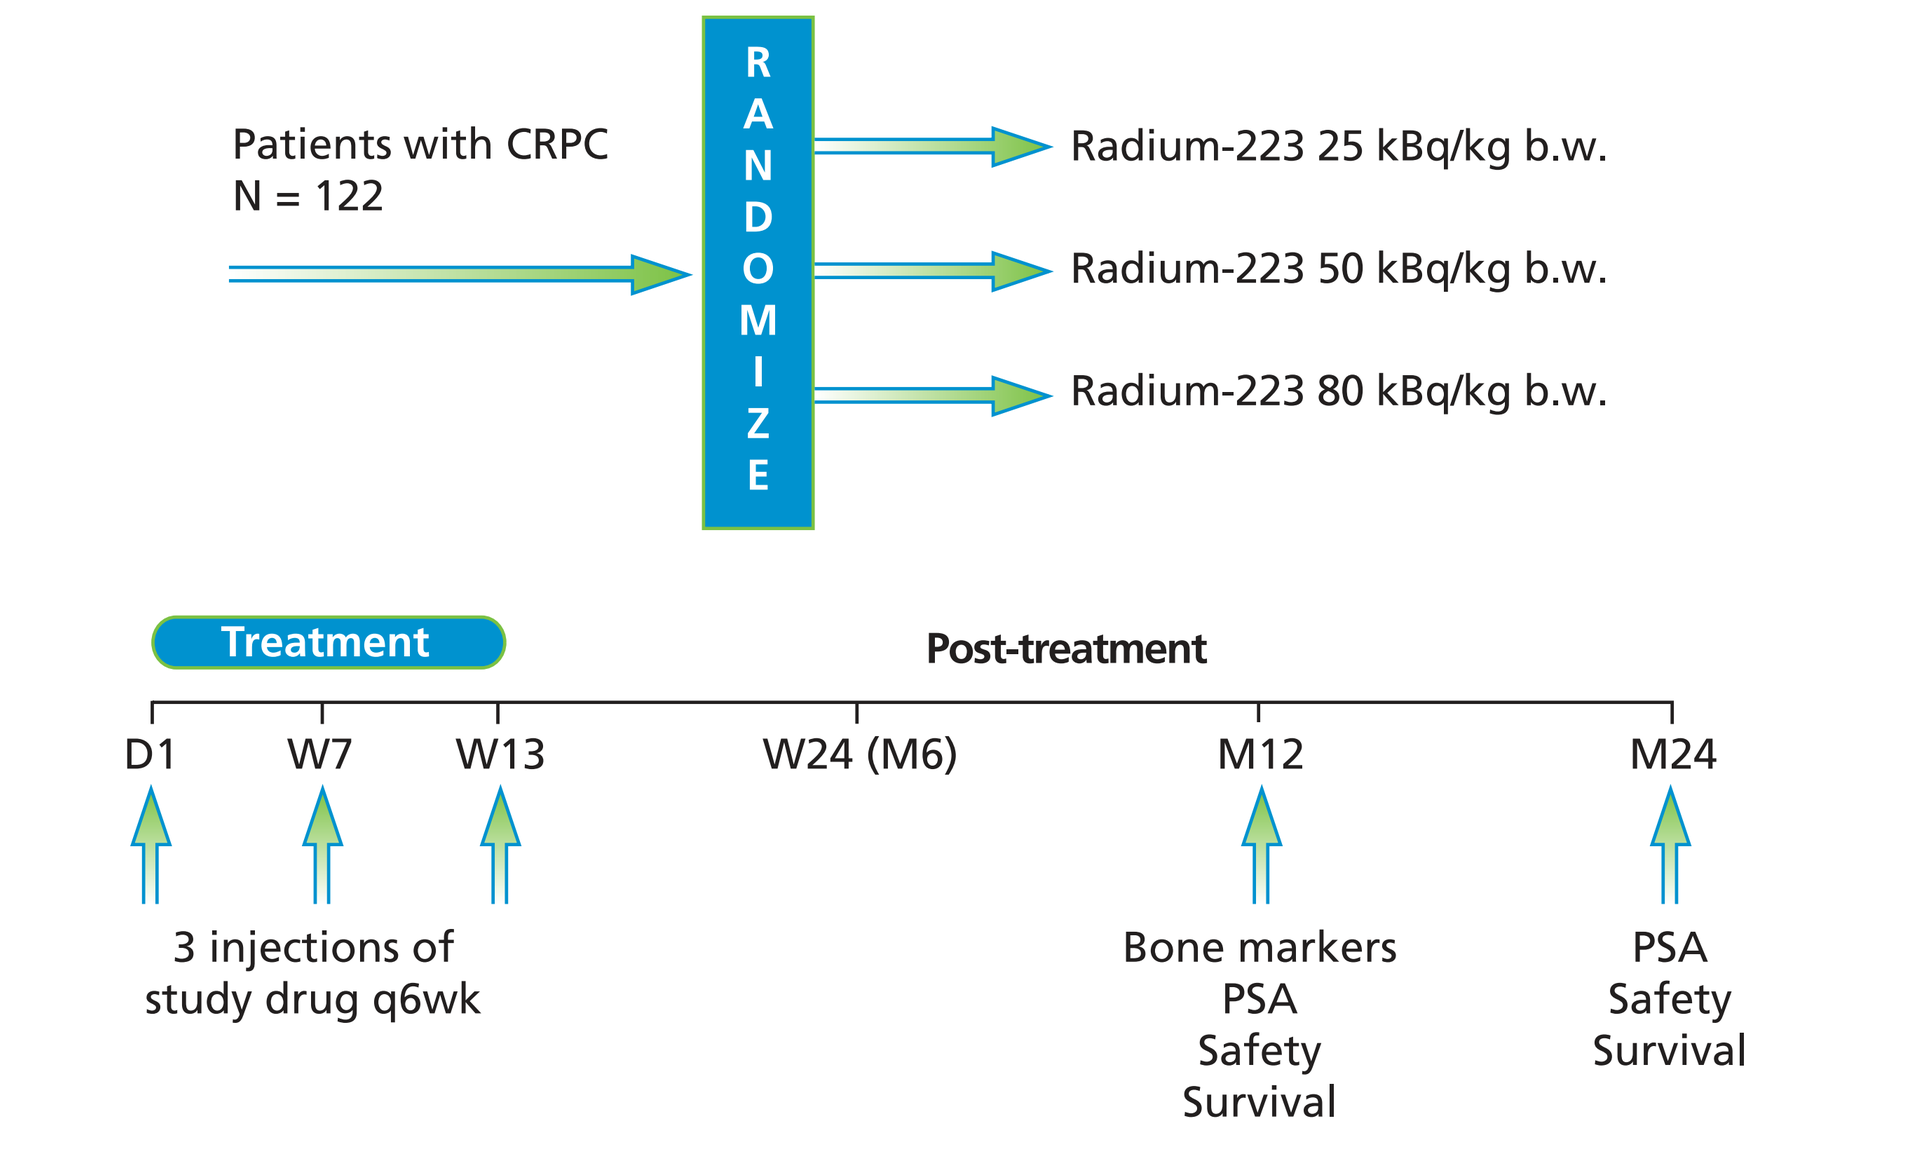
Patients with CRPC
N = 122
RANDOMIZE
Radium-223 25 kBq/kg b.w.
Radium-223 50 kBq/kg b.w.
Radium-223 80 kBq/kg b.w.
Treatment
Post-treatment
D1
W7
W13
W24 (M6)
M12
M24
3 injections of study drug q6wk
Bone markers
PSA
Safety
Survival
PSA
Safety
Survival

RESULTS

BC1-02 Study

24-Month Overall Survival

- Radium-223 improved OS by 4.5 months versus placebo when added to standard of care in patients with CRPC and bone metastases (65 vs 46 wk, respectively; HR 2.10 [1.14-3.88]; $P = .017$) (**Figure 3**)[5]
- OS in the BC1-02 study provides the context for evaluating ad hoc analyses comparing ALP normalization with OS

Figure 3. Overall survival of all randomly assigned patients in the BC1-02 study

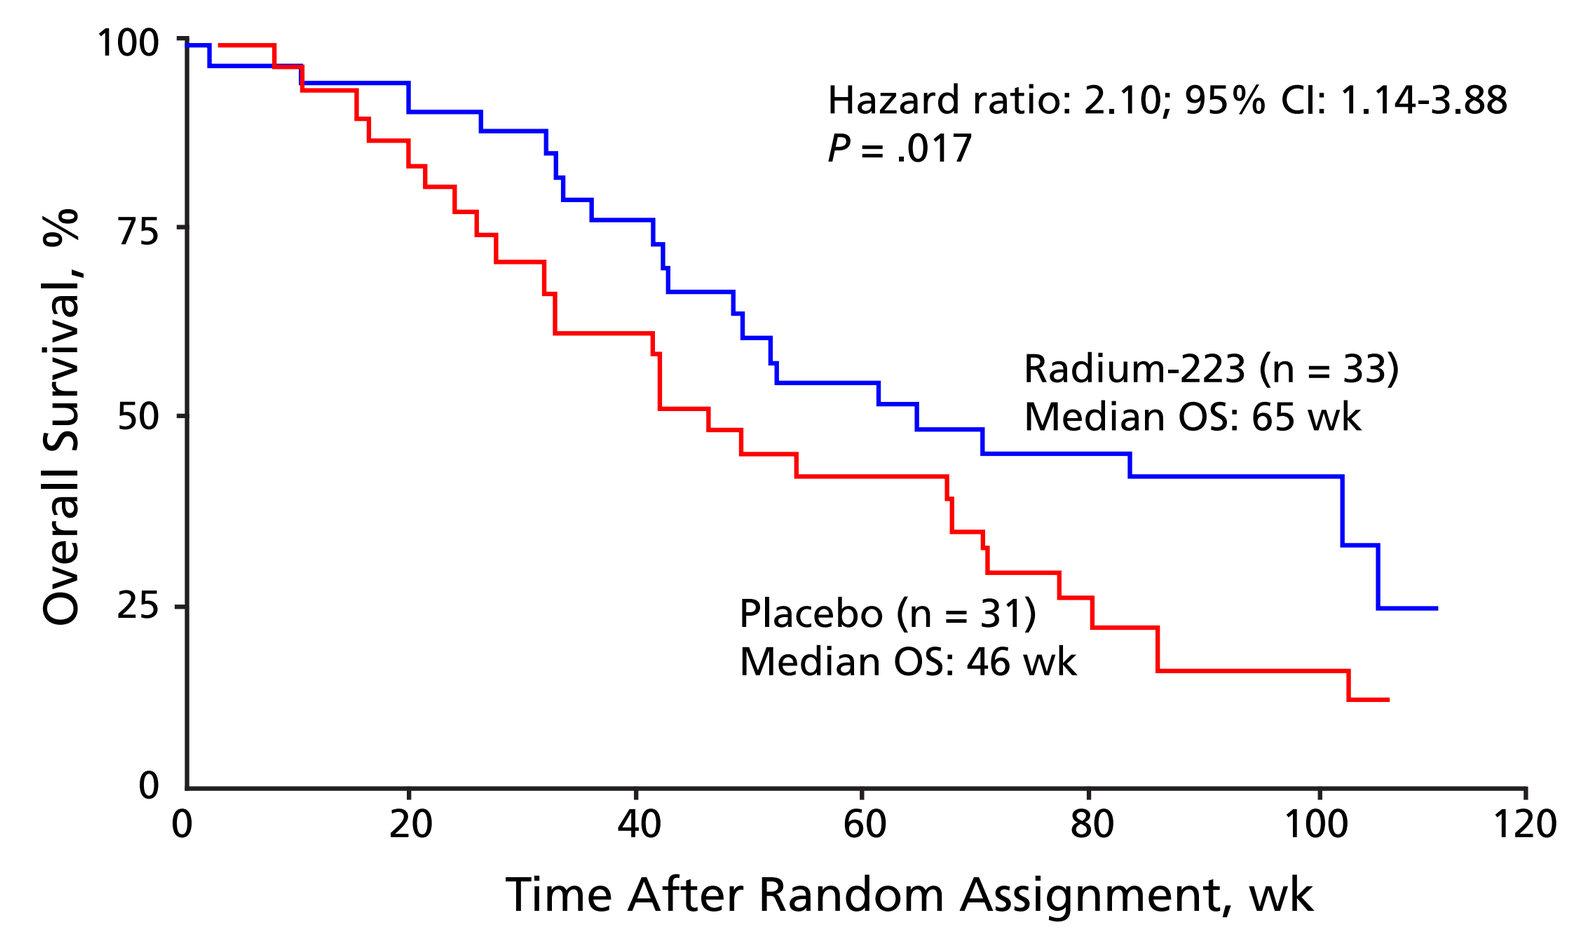
Hazard ratio: 2.10; 95% CI: 1.14-3.88
P = .017
Radium-223 (n = 33)
Median OS: 65 wk
Placebo (n = 31)
Median OS: 46 wk
Overall Survival, %
Time After Random Assignment, wk

ALP Normalization and Overall Survival

- In total, 54/64 (84%) patients enrolled in the BC1-02 study had an above-normal baseline total ALP value. In the radium-223 group, 26/33 (79%) patients had an above-normal baseline total ALP value versus 28/31 (90%) patients in the placebo group
- Of the 54 patients with an elevated baseline total ALP value, 2 had no postbaseline value; thus the analyses of normalization were based on 52 patients
- After treatment, 13/52 patients (25%) had normalized ALP values; however, there was a large treatment effect with normalized ALP values in 12 (46%) patients in the radium-223 group versus only 1 (4%) patient in the placebo group
- Median OS for the 13 patients with, and the 39 patients without, ALP normalization was 102 weeks and 43 weeks, respectively (log-rank $P < .001$) (**Table 1**)
- ALP normalization was associated with a significant improvement in OS for all patients with elevated baseline ALP values (**Figure 4**). This statistically significant improvement in OS was maintained in a separate analysis including only patients in the radium-223 group (**Figure 5**)

Table 1. Number of deaths and median OS for normalized versus nonnormalized patients in the BC1-02 study

	N	Deaths	Median OS, wk (Range)	*P* Value
All Patients				
Nonnormalized	39	37	42.6 (33-65.3)	< .001
Normalized	13	8	102.4 (52.7-NA)	
Radium-223 Patients				
Nonnormalized	14	14	42.5 (32.9-71)	< .001
Normalized	12	8	102.1 (52.7-NA)	

NA = not available.

Figure 4. Overall survival of all patients with normalized versus nonnormalized baseline ALP values in the BC1-02 study

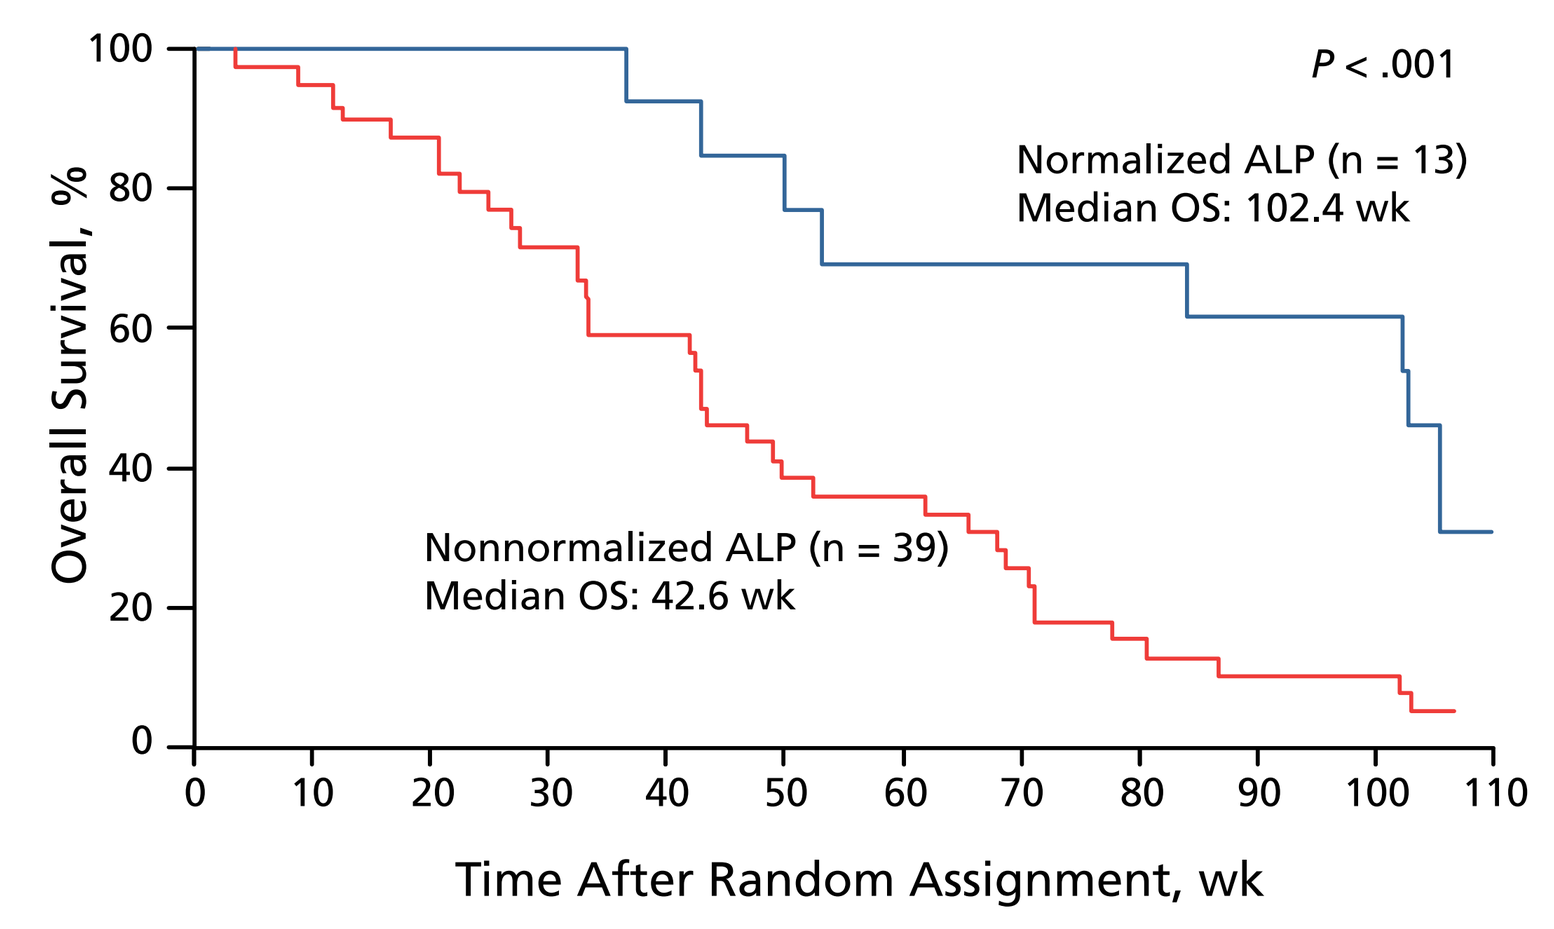
P < .001
Normalized ALP (n = 13)
Median OS: 102.4 wk
Nonnormalized ALP (n = 39)
Median OS: 42.6 wk
Overall Survival, %
Time After Random Assignment, wk

Figure 5. Overall survival of patients in the radium-223 group of the BC1-02 study who had normalized versus nonnormalized baseline ALP values

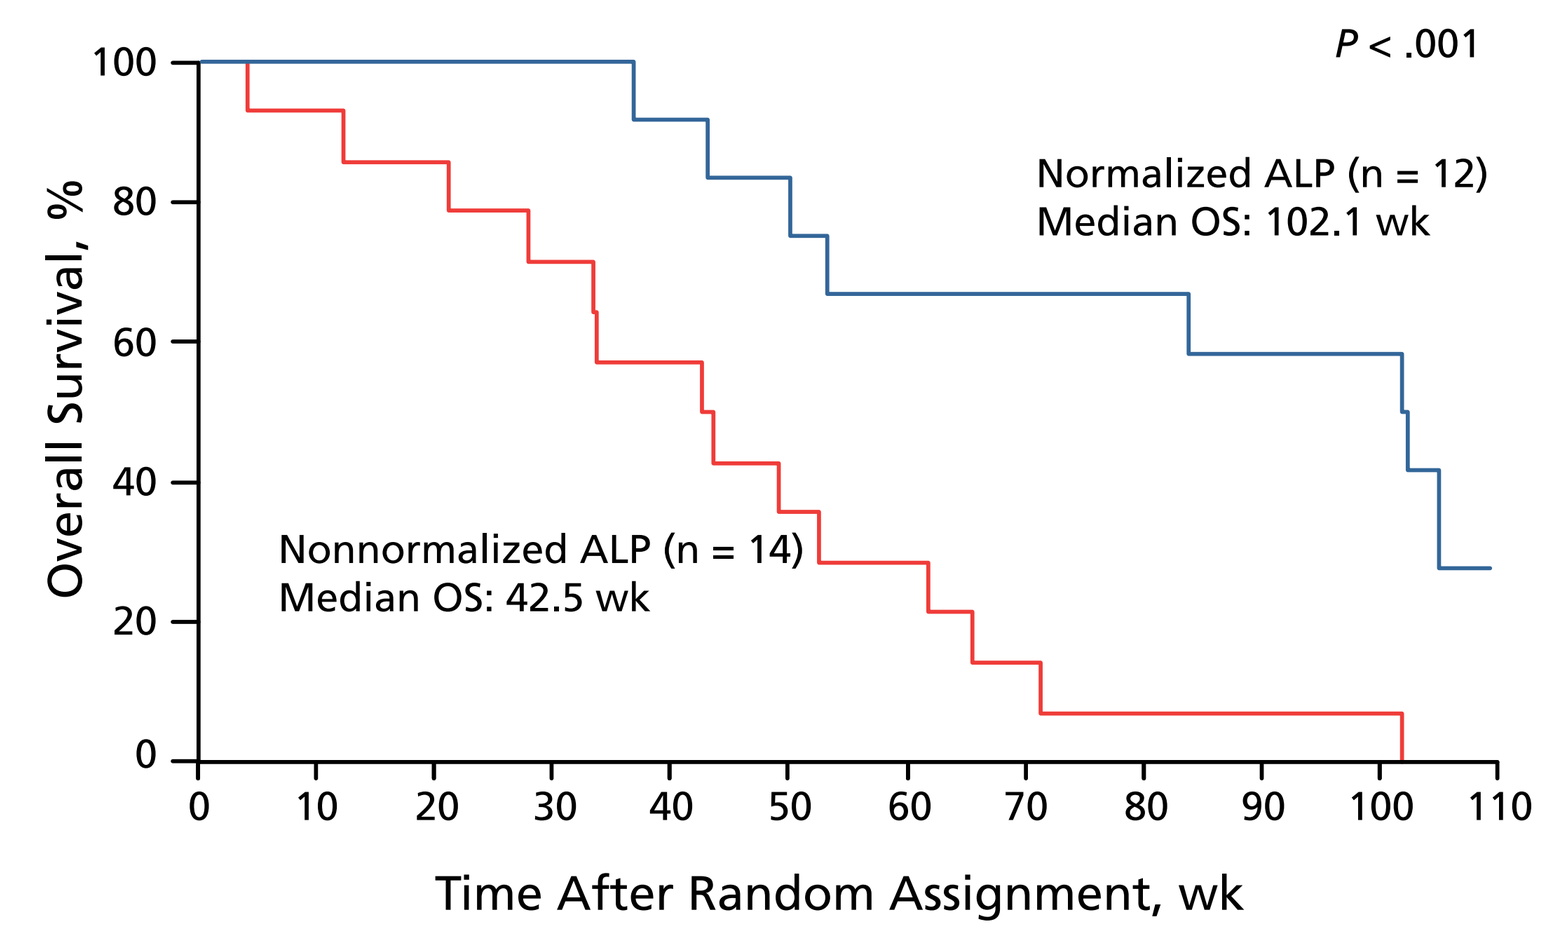
P < .001
Normalized ALP (n = 12)
Median OS: 102.1 wk
Nonnormalized ALP (n = 14)
Median OS: 42.5 wk
Overall Survival, %
Time After Random Assignment, wk

BC1-04 Study

ALP Normalization and Overall Survival

- A total of 75/122 (61%) patients in the BC1-04 study had an above-normal baseline total ALP value
- **After treatment with radium-223, ALP normalization was seen in 25/75 (33%) cases: 5/29 (17%) in the 25 kBq/kg group, 10/25 (40%) with 50 kBq/kg, and 10/21 (48%) with 80 kBq/kg, indicating a dose-response relationship**
- Median survivals for the 25 patients with, and the 50 patients without, ALP normalization were 102 weeks and 58 weeks, respectively (log-rank $P = .0086$) (**Table 2**)
- ALP normalization was associated with a significant improvement in OS for patients in the BC1-04 study (**Figure 6**)

Table 2. Number of deaths and median OS for normalized versus nonnormalized patients in the BC1-04 study

	N	Deaths	Median OS, wk (Range)	*P* Value
All Patients				
Nonnormalized	50	36	57.6 (38.1-73)	.0086
Normalized	25	13	101.7 (73.3-NA)	

NA = not available.

Figure 6. Overall survival of patients in the BC1-04 study who had normalized versus nonnormalized baseline ALP values

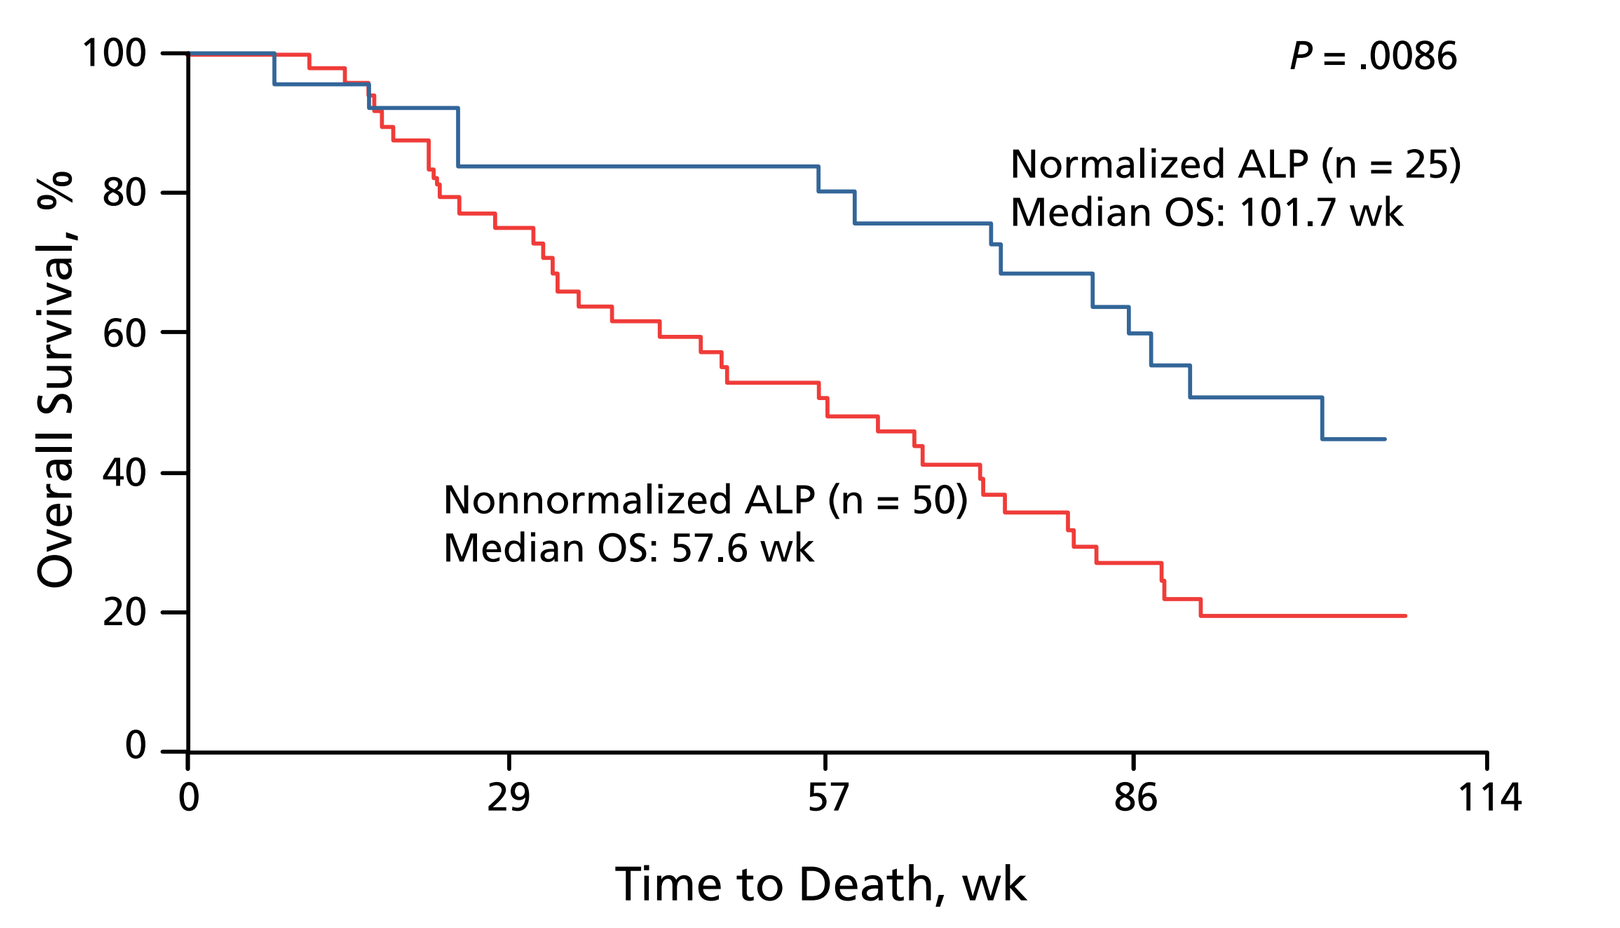
P = .0086
Normalized ALP (n = 25)
Median OS: 101.7 wk
Nonnormalized ALP (n = 50)
Median OS: 57.6 wk
Overall Survival, %
Time to Death, wk

Safety

- The overall AE profile was consistent with that expected for patients with advanced cancer and bone metastases, and was characterized by gastrointestinal events (eg, nausea, vomiting, diarrhea, and constipation), bone pain, fatigue, and anemia. No signs of renal or hepatic toxicity were observed
- In the placebo-controlled phase II study (BC1-02), there were more AEs and serious AEs in the placebo group than in the radium-223 group (**Table 3**)

Table 3. Summary of adverse events in the placebo-controlled phase II study (BC1-02)

	Radium-223 (n = 33)	Placebo (n = 31)
Number of adverse events	155	174
Number of patients with at least 1 adverse event	31	30
Number of serious adverse events (SAEs)	12	19
Number of patients with at least 1 SAE	8	14

- In the BC1-04 study, 145/551 AEs (26%) were considered treatment related: 33, 57, and 55 AEs, respectively, in the 25 kBq/kg, 50 kBq/kg, and 80 kBq/kg dose groups. Treatment-related AEs were primarily CTC grade 1 or 2; 10 events were grade 3, and 2 events were grade 4
- In the BC1-02 and BC1-04 studies, mild reversible neutropenia was observed, with neutrophil counts returning to baseline after completion of treatment

CONCLUSIONS

- In patients with CRPC and bone metastases treated with radium-223, ALP normalization was associated with a significant improvement in OS
- In these phase II clinical trials of almost 200 patients with CRPC and bone metastases, radium-223 demonstrated a highly tolerable safety profile characterized by generally manageable, low-grade AEs and a low propensity for hematologic events
- A randomized phase III survival study, ALSYMPCA, is ongoing worldwide,[10] and a phase I combination study with docetaxel was recently initiated (BC1-10)[11]

REFERENCES

1. Lange and Vessella. *Cancer Metastasis Rev*. 1999;17:331-336.
2. Henriksen et al. *Cancer Res*. 2002;62:3120-3125.
3. Nilsson et al. *Lancet Oncol*. 2007;8:587-594.
4. Data on file.
5. Nilsson et al. *Eur J Cancer Suppl*. 2009;7:411. Abstract P-7018.
6. Nilsson et al. *Eur J Cancer Suppl*. 2009;7:409. Abstract P-7011.
7. Parker et al. *Eur J Cancer Suppl*. 2009;7:406. Abstract O-7003.
8. Robinson et al. *J Urol*. 2008;179:117-123.
9. Sonpavde et al. *Urol Oncol*. 2010;Sep 29 [Epub ahead of print].
10. ClinicalTrials.gov Registration Number: NCT00699751 http://www.clinicaltrials.gov/ct2/show/NCT00699751.
11. ClinicalTrials.gov Registration Number: NCT01106352 http://www.clinicaltrials.gov/ct2/show/NCT01106352.