Abstract 4678

Radium-223 Chloride, a First-in-Class Alpha-Pharmaceutical With a Benign Safety Profile for Patients With Castration-Resistant Prostate Cancer (CRPC) and Bone Metastases: Combined Analysis of Phase I and II Clinical Trials

S. Nilsson,[1] C. Parker,[2] I. Haugen,[3] A. Lokna,[3] A.K. Aksnes,[3] B. Bolstad,[3] G. O'Bryan-Tear,[3] Ø.S. Bruland[4]

[1]*Karolinska University Hospital, Stockholm, Sweden;* [2]*Royal Marsden Hospital, Surrey, UK;* [3]*Algeta ASA, Oslo, Norway;* [4]*University of Oslo Norwegian Radium Hospital, Oslo, Norway*

BACKGROUND

- Bone metastases are the main cause of disability and death in patients with castration-resistant prostate cancer (CRPC)[1]
- Radium-223 chloride (Alpharadin) is the first-in-class alpha-pharmaceutical which has a potent and highly targeted antitumor effect on bone metastases,[2] and a highly tolerable side effect profile[3]
- Bone-targeted radium-223 emits alpha-particles with an ultra-short range of 2-10 cell diameters, generating highly localized and intense radiation zones.[2] The high-energy alpha-particle radiation induces lethal double-stranded DNA breaks[4] resulting in a potent and highly localized cytotoxic effect in the target areas containing metastatic cancer cells. The short path length of the alpha-particles also ensures that toxicity to adjacent healthy tissue and particularly the bone marrow is kept to a minimum[2]
- In the placebo-controlled phase II study (BC1-02), radium-223 showed a statistically significant improvement in overall survival compared to placebo,[3,5] and a consistent improvement in disease-related biomarkers[3,6,7] and pain[6]
- A combined evaluation was conducted to assess the overall safety and tolerability profile of radium-223 among the 292 patients with bone metastases and mainly CRPC who participated in phase I and II clinical trials

METHODS AND STUDY DESIGN

- A total of 292 patients with mainly CRPC and bone metastases received radium-223 in 2 open-label phase I trials (37 patients), 2 double-blind, dose-response phase II trials (222 patients), and 1 double-blind, placebo-controlled phase II trial (33 patients received radium-223 and 31 received placebo) (Figure 1)
- Radium-223 was administered at doses of 5 to 250 kBq/kg, given as either single or repeated injections
- The trials assessed safety, efficacy (changes in prostate-specific antigen [PSA], bone alkaline phosphatase [ALP], pain, and survival), biodistribution, and dosimetry
- Adverse events data were compiled and analyzed descriptively

Figure 1. Phase I and II studies

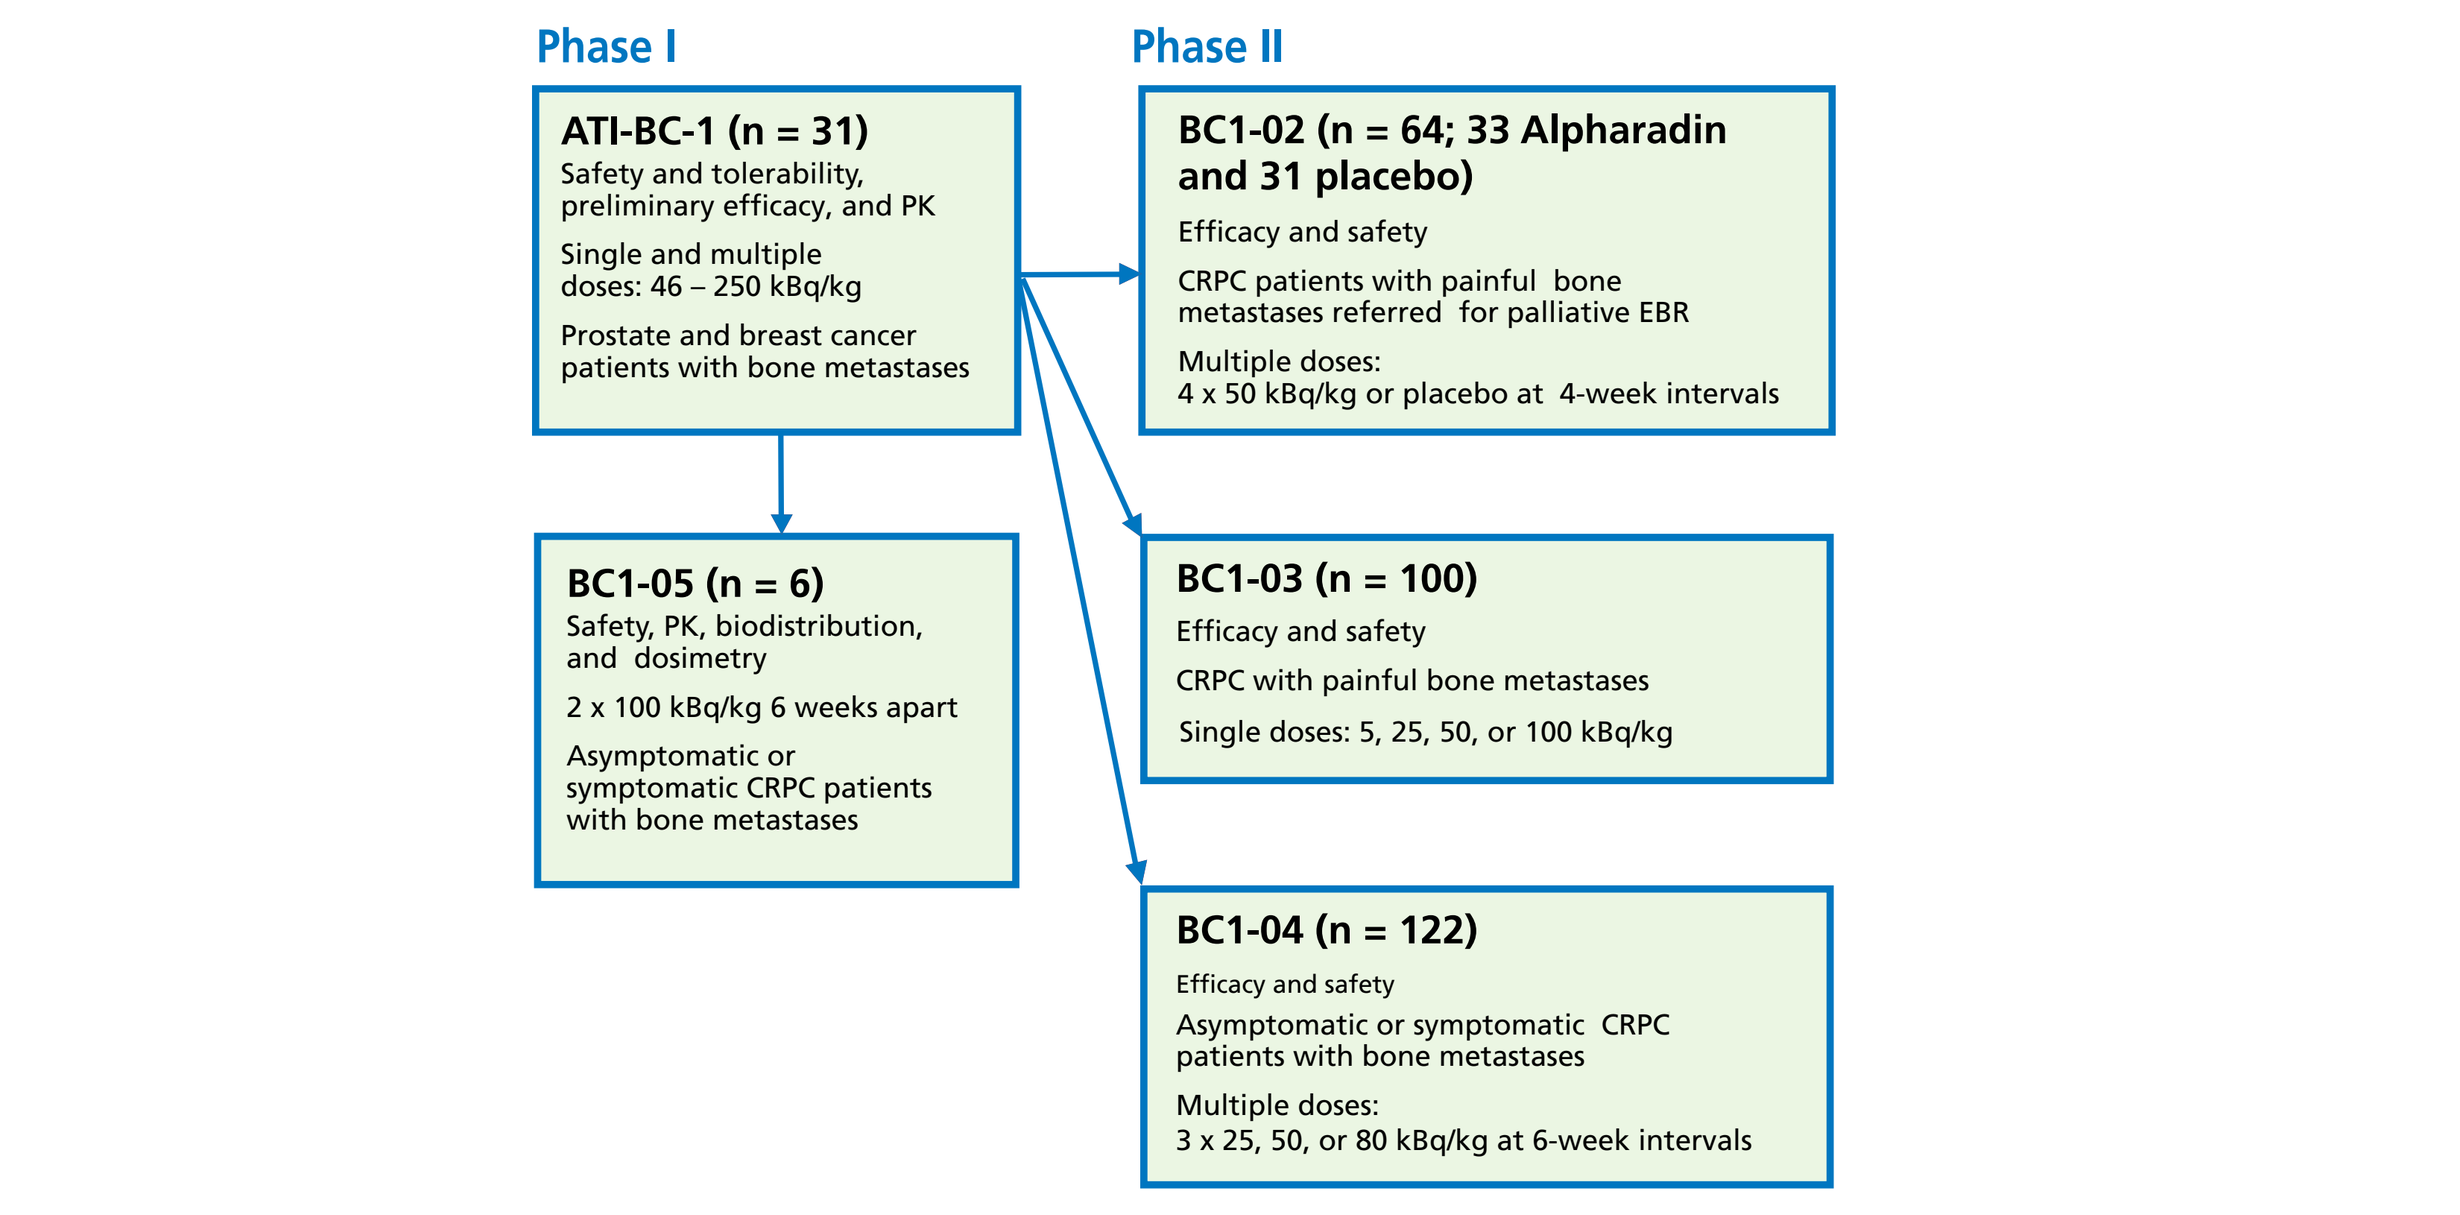
Phase I
Phase II
ATI-BC-1 (n = 31)
Safety and tolerability, preliminary efficacy, and PK
Single and multiple doses: 46 – 250 kBq/kg
Prostate and breast cancer patients with bone metastases
BC1-05 (n = 6)
Safety, PK, biodistribution, and dosimetry
2 x 100 kBq/kg 6 weeks apart
Asymptomatic or symptomatic CRPC patients with bone metastases
BC1-02 (n = 64; 33 Alpharadin and 31 placebo)
Efficacy and safety
CRPC patients with painful bone metastases referred for palliative EBR
Multiple doses:
4 x 50 kBq/kg or placebo at 4-week intervals
BC1-03 (n = 100)
Efficacy and safety
CRPC with painful bone metastases
Single doses: 5, 25, 50, or 100 kBq/kg
BC1-04 (n = 122)
Efficacy and safety
Asymptomatic or symptomatic CRPC patients with bone metastases
Multiple doses:
3 x 25, 50, or 80 kBq/kg at 6-week intervals

RESULTS

Blood clearance, excretion, and uptake

- Radium-223 was rapidly eliminated from blood; more than 75% of the activity had left the blood and plasma at 15 minutes after injection. Only 4% ± 1% (range 2%-6%) of the activity remained in the blood at 4 hours after injection
- Radium-223 exhibited a high initial uptake into the bone (44%-77% of the injected activity at 4 h) or small intestine (19%-69% of the injected activity at 4 h). Fecal excretion is the main route of elimination from the body
- There was no hepatobiliary excretion, and very little or no uptake into the kidneys, liver, or other internal organs

Adverse events and serious adverse events

- In the placebo-controlled phase II study (BC1-02) there were more adverse events and serious adverse events in the placebo group than in the radium-223 group (Table 1)
- The most frequently reported adverse events (> 15% of patients) across phase I and II studies for radium-223 and placebo are summarized in Table 2

Table 1. Summary of adverse events in the placebo-controlled phase II study

	Radium-223 (n = 33)	Placebo (n = 31)
Number of adverse events	155	174
Number of patients with at least 1 adverse event	31	30
Number of serious adverse events (SAEs)	12	19
Number of patients with at least 1 SAE	8	14

Table 2. Most frequent adverse events reported for radium-223 or placebo in phase I and II clinical trials

	Number (%) of patients	
Adverse event	Radium-223 n = 292	Placebo* n = 31
Nausea	97 (33)	10 (32)
Bone pain	89 (30)	16 (52)
Fatigue	76 (26)	7 (23)
Diarrhea	77 (26)	10 (32)
Anemia	71 (24)	7 (23)
Constipation	60 (21)	2 (7)
Vomiting	59 (20)	6 (19)

*Placebo events recorded in the BC1-02 clinical trial.

- The overall adverse event profile was consistent with that expected for patients undergoing treatment for advanced cancer with bone metastases, and was characterized by gastrointestinal events (nausea, vomiting, diarrhea, and constipation), bone pain, fatigue, and anemia. No signs of renal or hepatic toxicity were observed
- Serious adverse events were reported in 34% of patients (98/292). Those recorded in more than 2% of the patients were bone pain (5%), anemia (4%), spinal cord compression (3%), and disease progression (2.7%)
- In the placebo arm of the BC1-02 study, bone pain was the only serious adverse event reported in more than 1 patient (3 patients/10%). Among other events, anemia and spinal cord compression were reported by 1 patient (3%)

Hematologic adverse events

- In the first phase I study (ATI-BC-1), 5 patients in each dose group received doses of 46, 93, 163, 213, and 250 kBq/kg body weight (b.w.) (dose groups 1-5) as a single injection
- Mild, transient myelosuppression of platelets and neutrophils was seen (Figure 2). Platelet count reductions were restricted to NCI CTC grade 1, while 2 patients experienced grade 3 reductions in neutrophil counts (1 each at radium-223 dosages of 163 and 250 kBq/kg b.w.)

Figure 2. Mild, transient neutropenia observed in phase I (ATI-BC-1)

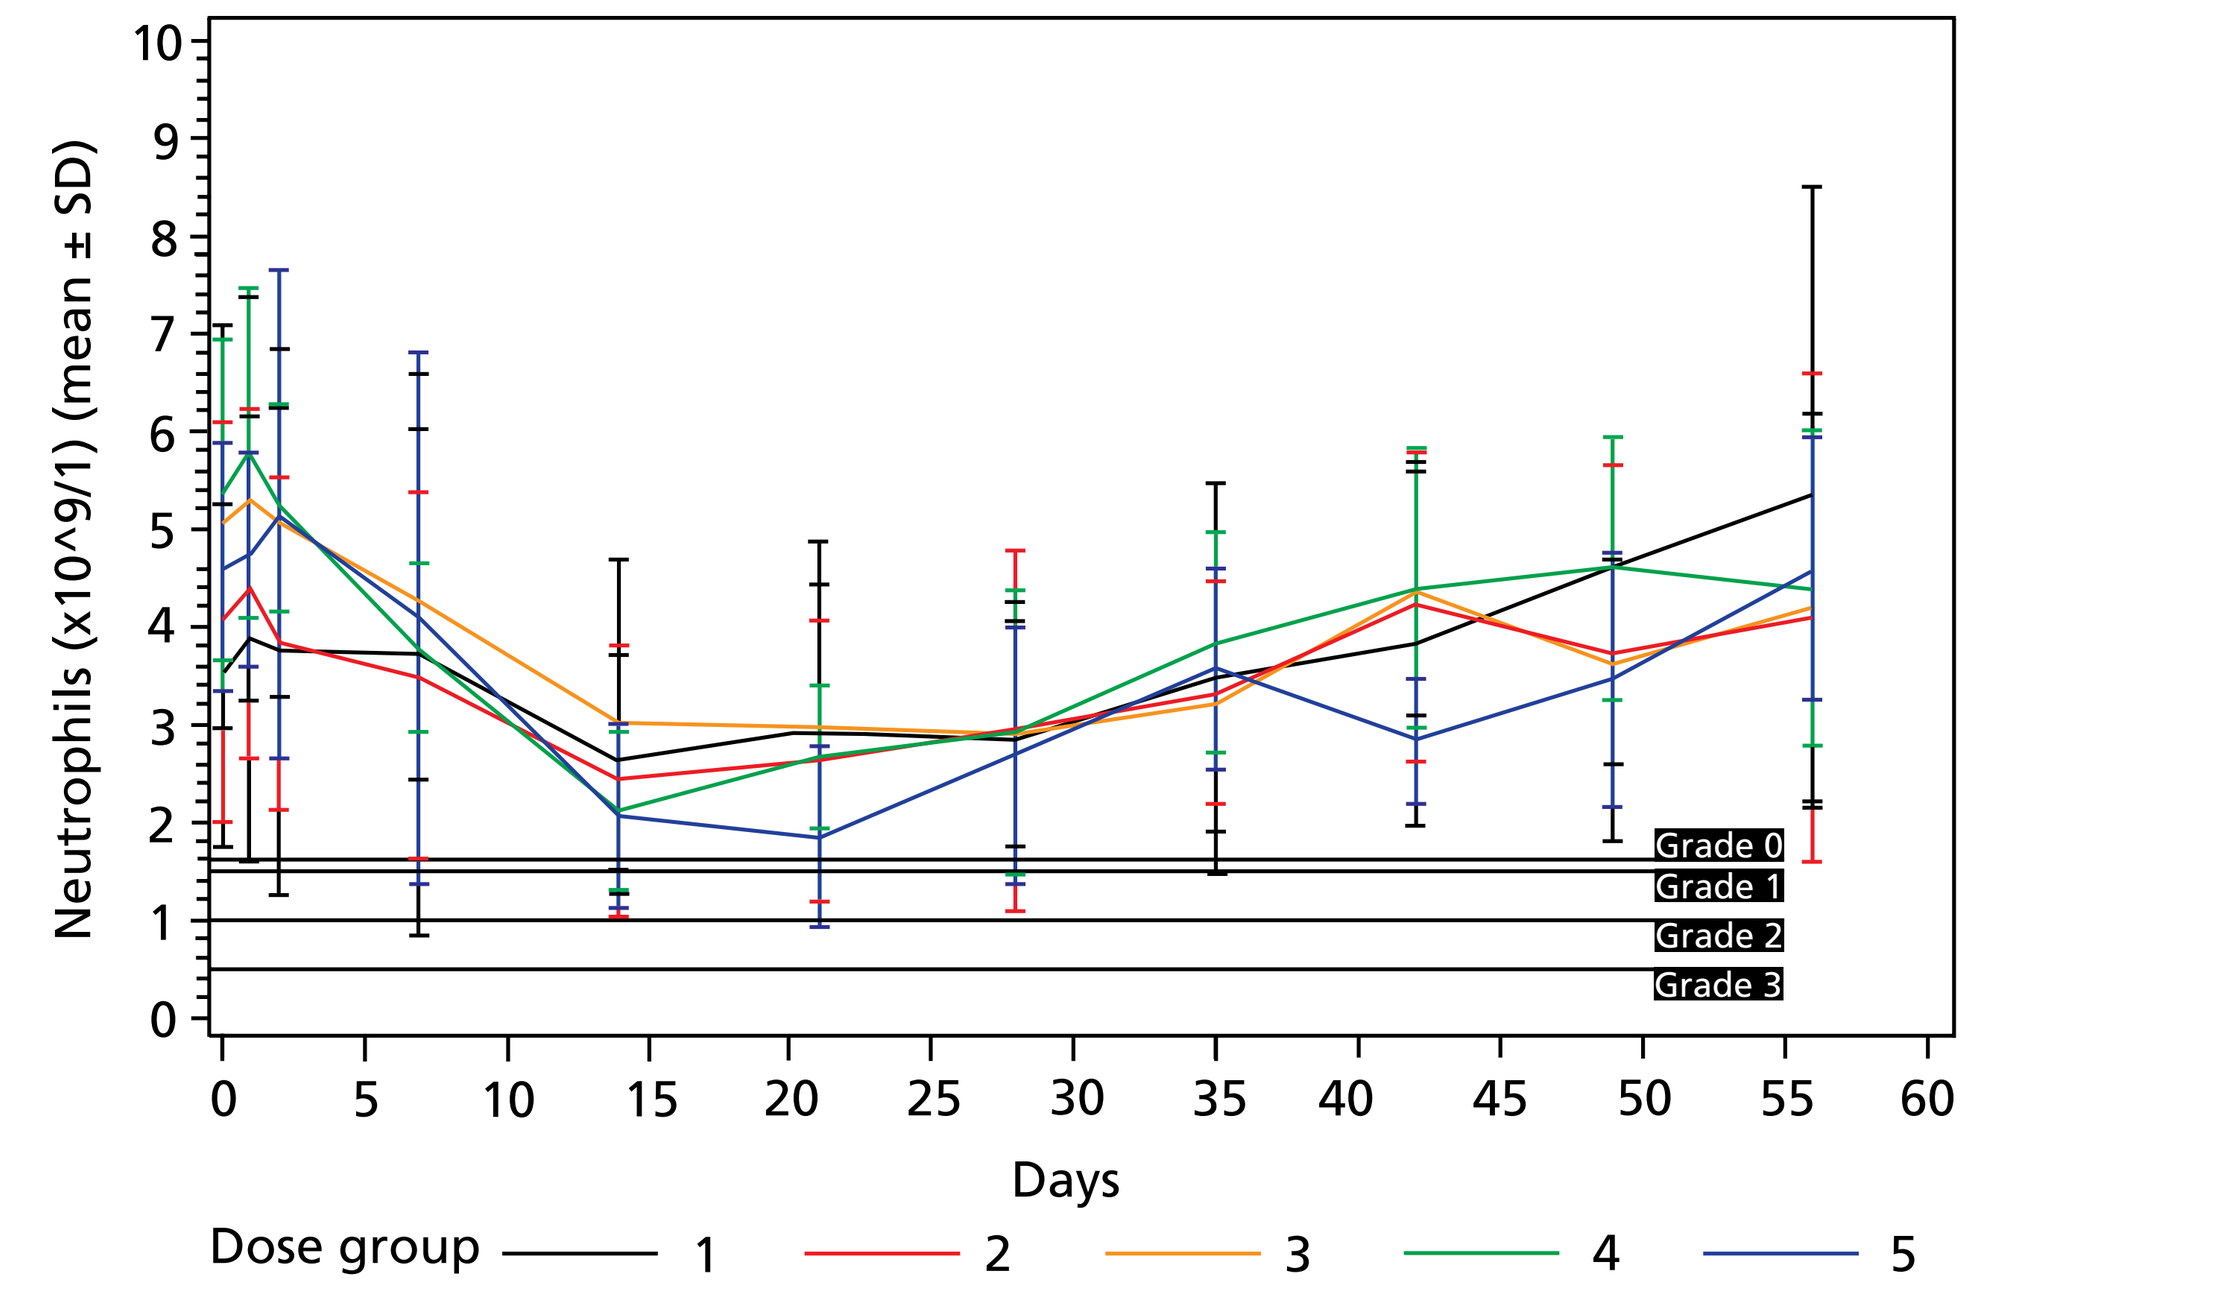
Neutrophils (x10^9/1) (mean ± SD)
0
1
2
3
4
5
6
7
8
9
10
Grade 0
Grade 1
Grade 2
Grade 3
0
5
10
15
20
25
30
35
40
45
50
55
60
Days
Dose group
1
2
3
4
5

- In the placebo-controlled phase II study (BC1-02), no patients in the radium-223 group experienced grade 4 hematologic adverse events. One patient in the placebo group experienced grade 4 anemia (Table 3)
- No more than 1% of the 292 patients across all studies experienced NCI CTC grade 4 hematologic adverse events during the study period (Table 4)
- 4.8% of patients experienced NCI CTC grade 3 toxicity for hemoglobin, and less than 3% experienced grade 3 toxicity for platelets, neutrophils, or white blood cells (WBC)

Table 3. Hematologic toxicity from the placebo-controlled phase II study (BC1-02)

Worst grade for hematologic toxic effects by patient during treatment: 16 weeks, 4 injections of 50 kBq/kg at 4-week intervals*

	Radium-223 (n = 33)				Placebo (n = 31)			
NCI CTC Grade	1	2	3	4	1	2	3	4
Platelets	6	0	0	0	4	0	1	0
Neutrophils	5	2	1	0	0	0	0	0
WBC	9	1	1	0	3	0	0	0
Hemoglobin	26	4	1	0	19	6	0	1

*Data are number of patients.

Table 4. Hematologic toxicity across all studies (n = 292)

Worst grade for hematologic toxic effects by patient during treatment, all patients receiving radium-223 in any study and any dose (n = 292)

	Radium-223 (n = 292)			
NCI CTC Grade	1	2	3	4
Platelets	59 (20%)	6 (2%)	6 (2%)	3 (1%)
Neutrophils	57 (20%)	32 (11%)	5 (1.7%)	2 (0.7%)
WBC	53 (18%)	41 (14%)	8 (2.7%)	0
Hemoglobin	149 (51%)	72 (25%)	14 (4.8%)	3 (1%)

Long-term patient follow-up

- The patients have been followed for up to 3 years (ATI-BC-1: 3 years; BC1-02, BC1-03, and BC1-04: 2 years; BC1-05: 1 year). Potential long-term toxicity has been assessed and specified as acute myeloid leukemia (AML), myelodysplastic syndrome (MDS), aplastic anemia, and primary bone cancer. No patients were given any of these diagnoses
- No apparent difference in hematologic parameters between the placebo and the radium-223 group was observed in the follow-up period in the BC1-02 study

EFFICACY EVALUATIONS

- Efficacy evaluations showed consistent improvement in disease-related biomarkers (bone and PSA) and pain[3,6,7]
- In the placebo-controlled study, radium-223 improved overall survival by 4.5 months versus placebo when added to standard of care (65 vs 46 wk, respectively; HR 2.10 [1.14-3.88]; P = .017)[5]

CONCLUSIONS

- This combined analysis of phase I and II clinical trials of almost 300 patients with bone metastases and (mainly) CRPC provides valuable insights into the safety and efficacy profile of radium-223
- Radium-223 has shown a highly tolerable safety profile characterized by generally manageable, low-grade toxicities and a low propensity for hematologic events
- These findings were accompanied by improvements in disease-related biomarkers and pain, and a significant improvement in survival
- A randomized phase III survival study, ALSYMPCA, is ongoing worldwide

REFERENCES

1. Lange and Vasella. *Cancer Metastasis Rev*. 1999;17:331-336.
2. Henriksen et al. *Cancer Res*. 2002;62:3120-3125.
3. Nilsson et al. *Lancet Oncol*. 2007;8:587-594.
4. Data on file.
5. Nilsson et al. *Eur J Cancer Suppl*. 2009;7:411. Abstract P-7018.
6. Nilsson et al. *Eur J Cancer Suppl*. 2009;7:409. Abstract P-7011.
7. Parker et al. *Eur J Cancer Suppl*. 2009;7:406. Abstract O-7003.